Enhanced Antigen Capture, Antigen-Presenting Cell-Like Function, and Cytotoxic Responses With Chimeric Engulfment Receptor T Cells (CER T Cells)

Poster 207

Daniel Corey[1], Sunil Thomas[1], Brandon Cieniewicz[1], Linh Nguyen[1], Jared Clever[1], Josephine Brysting[1], Remus Vezan[1], John M. Rossi[1], Kurt Diem[2], Lei Jin[2], Lawrence Corey[2]

[1]CERo Therapeutics, San Francisco, CA, USA; [2]Vaccine and Infectious Disease Division, Fred Hutchinson Cancer Research Center, Seattle, WA, USA

INTRODUCTION

- Activated T cells have limited soluble antigen-presenting capabilities due to inefficient antigen capture[1]
- T cells were engineered to express novel chimeric engulfment receptors (CERs) containing an extracellular human T-cell immunoglobulin and mucin domain containing-4 (Tim-4) phagocytic receptor that recognizes phosphatidylserine (PS)[2] fused to T-cell and/or macrophage/dendritic cell-derived signaling domains
 - CERs are designed to facilitate antigen capture, processing, and presentation, and impart target-dependent cytotoxic function when expressed in T cells
 - This combined function is hypothesized to improve tumor clearance and durability of response, making CER T-cell products ideal clinical candidates
 - Tumor specificity is imparted by inducing exposure of PS, which is normally restricted to the inner leaflet of the cell membrane, on the tumor cell surface using targeted therapies, such as a small molecule inhibitor

OBJECTIVES

- To design and manufacture novel CER T cells
- To characterize CER T-cell function in vitro

METHODS

- CER T cells containing the Tim-4 receptor fused to various transmembrane and intracellular signaling domains were generated using healthy donor T cells (**Figure 1**)
 - CER T-cell products were characterized on day 6 following transduction for expression of Tim-4, CD45RA, CCR7, and CD4/CD8 ratios, all measured by flow cytometry (FC) using commercially available fluorochrome-conjugated antibodies
- The induction of surface PS expression on REC-1 and JeKo-1 mantle cell lymphoma (MCL) cell lines treated with ibrutinib was measured by FC using histidine-tagged recombinant murine Tim-4; secondary detection was performed using an anti-histidine antibody
- Phagocytosis was assessed in co-culture assays of REC-1 cells with CER T cells
 - REC-1 cells were pretreated for 24 hours with 10 μM ibrutinib, labeled with pHrodo red, and cultured with untransduced T cells or CER T cells at 0.1 to 1 effector to target (E:T) ratios
 - The percentage of pHrodo red-positive cells was determined by FC after 16 or 40 hours of co-culture
 - The localization of tumor fragments in CER T-cell lysosomal compartments was assessed in a separate experiment with REC-1 cells labeled with pHrodo green; following co-culture, all cells were labeled with Lyso-tracker red
 - Visualization of engulfment was performed by fluorescence microscopy
 - 1-way ANOVA with Dunnett's post hoc test was performed to compare CER T cells with untransduced T cells
- CER T-cell cytotoxic function was evaluated using the Incucyte® live-cell analysis system in co-culture assays with REC-1 cells
 - REC-1 cells engineered to constitutively express mCherry (REC-1 mCherry) were pretreated with 10 μM ibrutinib or vehicle (20% 2-OH β-cyclodextrin) for 24 hours. Ibrutinib was washed off prior to co-culture with CER T cells
 - CER T cells (6-day expansion post-transduction) were co-cultured with ibrutinib-pretreated REC-1 cells at an E:T ratio of 1:1 in the presence of 0.5 μM ibrutinib or vehicle
 - Cytotoxicity is reflected by a decrease in fluorescence
- T-cell activation markers programmed death-1 (PD-1) and 4-1BB were evaluated on viable CD3+ T cells by FC using commercially available fluorochrome-conjugated antibodies
 - REC-1 mCherry cells were pretreated for 24 hours with 10 μM ibrutinib or vehicle; after wash out, co-cultures were initiated with 0.5 μM ibrutinib or vehicle
 - Assays were conducted 96 hours after initiation of co-culture of CER T cells with REC-1 cells
- Cytokine induction was assessed by multiparametric ELISA using conditioned supernatants harvested after 96 hours in the cytotoxicity assay described above
- Antigen-presenting cell (APC)-like function of CER T cells was assessed as follows (**Figure 2**)
 - Cell trace violet (CTV)-labeled E7 T-cell receptor (TCR) T cells were cultured for 6 days with untransduced T cells or CER T cells and JeKo-1 cells at a 1:2:2 ratio in the presence of a pool of 15-mer peptides with 11 amino acid overlap (100 ng of each peptide) derived from the HPV16 E7 protein
 - To demonstrate the role of major histocompatibility complex (MHC) class I antigen presentation, cultures as described above were incubated with a mouse anti-human leukocyte antigen (HLA) class I (A, B, and C) blocking antibody (clone W6/32) or matched mouse IgG2a antibodies for 4 days before analysis
 - Proliferation was measured by dye dilution of CTV

Figure 1. CER Design.

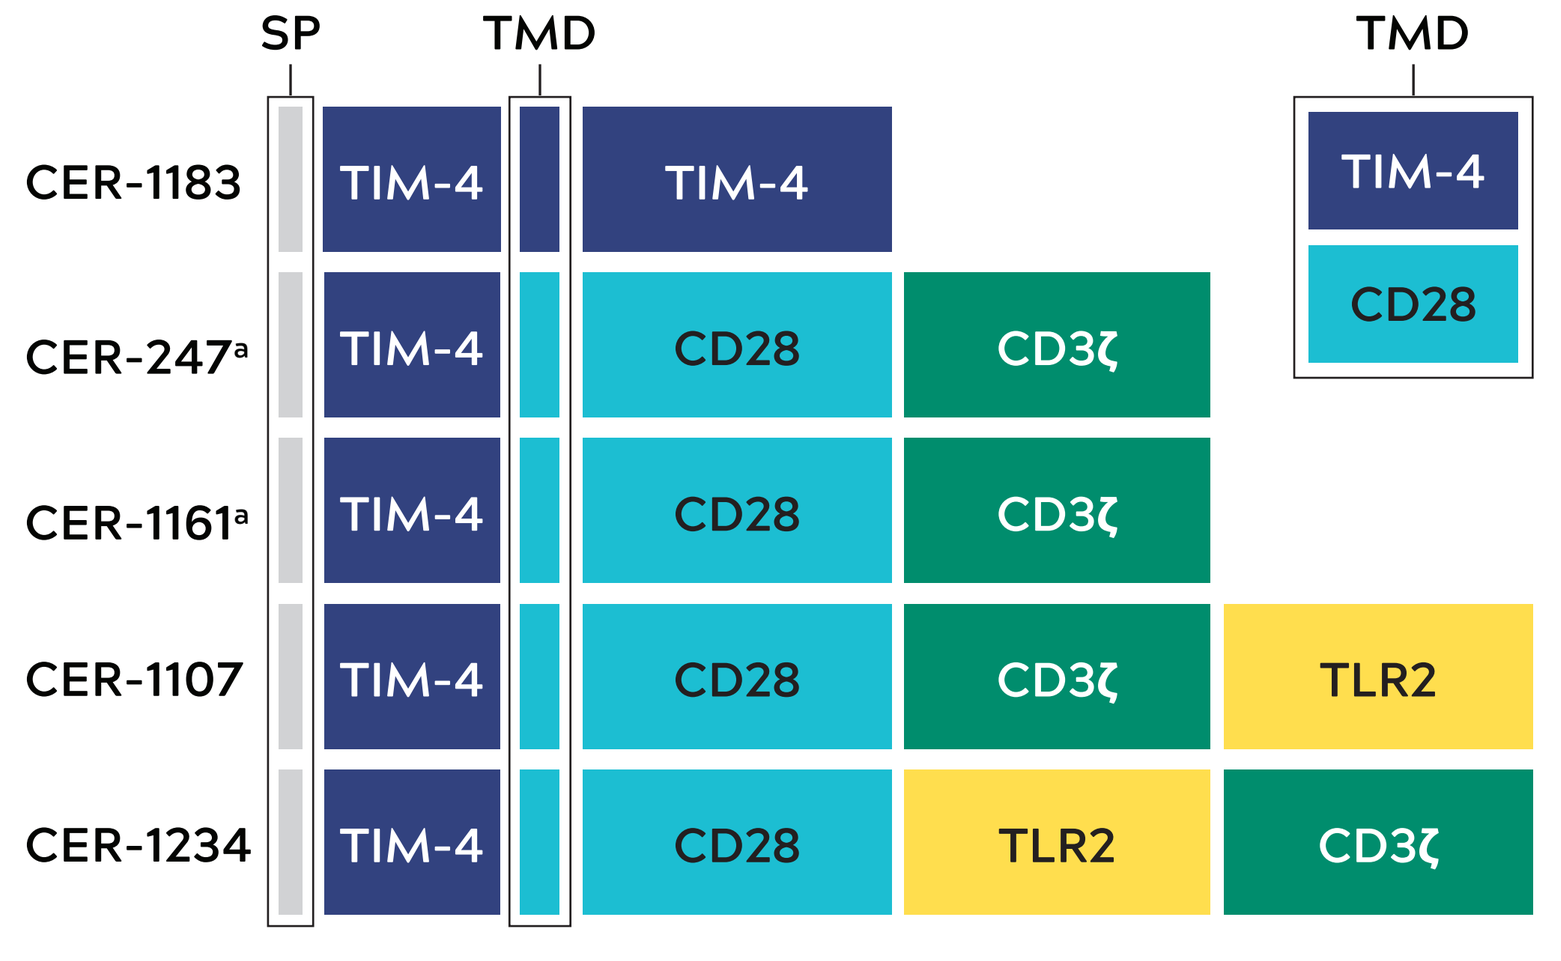
SP
TMD
TMD
CER-1183
CER-247[a]
CER-1161[a]
CER-1107
CER-1234
TIM-4
CD28
CD3ζ
TLR2

[a]CER-247 contains an EGFRt polypeptide tag.
CD28, cluster of differentiation 28; CD3ζ, T-cell surface glycoprotein CD3 zeta chain; CER, chimeric engulfment receptor; EGFRt, truncated form of epidermal growth factor receptor; SP, signaling peptide; Tim-4, T-cell immunoglobulin and mucin domain containing-4; TLR2, toll-like receptor-2; TMD, transmembrane domain.

Figure 2. Proliferation of E7 TCR T Cells Activated With E7 Peptide-Pulsed CER T Cells.

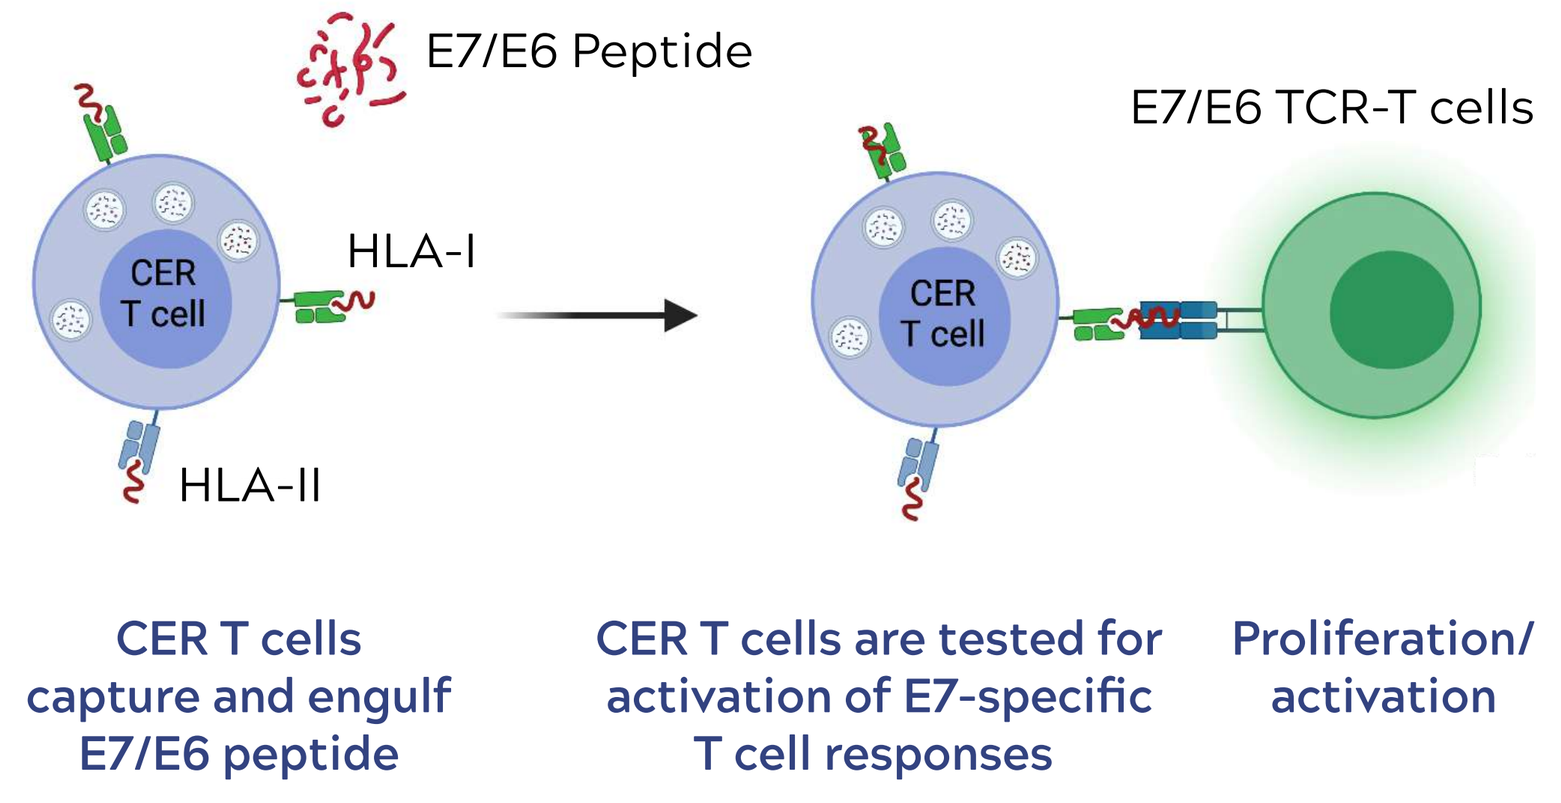
E7/E6 Peptide
E7/E6 TCR-T cells
CER T cell
HLA-I
HLA-II
CER T cells capture and engulf E7/E6 peptide
CER T cells are tested for activation of E7-specific T cell responses
Proliferation/ activation

CER, chimeric engulfment receptor; HLA, human leukocyte antigen; TCR, T-cell receptor.

RESULTS

DESIGN OF CER CONSTRUCTS AND CHARACTERISTICS OF CER T CELLS

- CERs were designed as single-chain chimeric proteins composed of different combinations of the following domains (**Figure 1**)
 - The Tim-4 extracellular domains that binds PS
 - The transmembrane domain of Tim-4 or CD28
 - The intracellular CD28 and CD3ζ signaling domains that serve as T-cell activators
 - The intracellular TLR2 signaling domain to enhance APC-like function
- CERs were expressed by T cells (**Table 1**)
 - High proportions of CER T cells express the engineered Tim-4 receptor
 - CD4/CD8 ratios were similar after transduction and expansion of CER T cells versus untransduced T cells
 - CER T cells exhibit a predominantly central memory and effector memory phenotype as measured by CCR7 and CD45RA

Table 1. CER T Cell Characteristics.[a]

Parameter	Untransduced	CER-1183	CER-1234
Tim-4⁺,%	Undetected	69.4	75.0
T-cell CD4/CD8 ratio	0.6	0.6	0.6
T-cell subsets, %			
CD45RA⁺/CCR7⁺ (naive)	11.8	13.8	14.6
CD45RA⁻/CCR7⁺ (central memory)	47.6	41.2	42.2
CD45RA⁻/CCR7⁻ (effector memory)	33.8	33.9	32.2
CD45RA⁺/CCR7⁻ (effector)	6.9	11.2	10.9

[a]On day 6 following transduction.
CD4, cluster of differentiation 4; CD8, cluster of differentiation 8; CER, chimeric engulfment receptor; Tim-4, T-cell immunoglobulin and mucin domain containing-4.

IBRUTINIB INDUCES PS EXPOSURE IN REC-1 AND JEKO-1 CELLS

- Ibrutinib (0.05 to 50 μM for 24 to 48 hours) induced PS exposure measured by FC (**Figure 3, A and B**)
 - REC-1 cells were more sensitive than JeKo-1 cells to ibrutinib-induced PS exposure

Figure 3. Induction of PS Exposure With Ibrutinib.
(**A**) PS exposure with ibrutinib (0.05 μM to 50 μM) at 24 hours in REC-1 and JeKo-1 cells. (**B**) PS exposure with ibrutinib (0.05 μM to 50 μM) at 48 hours in REC-1 and JeKo-1 cells.

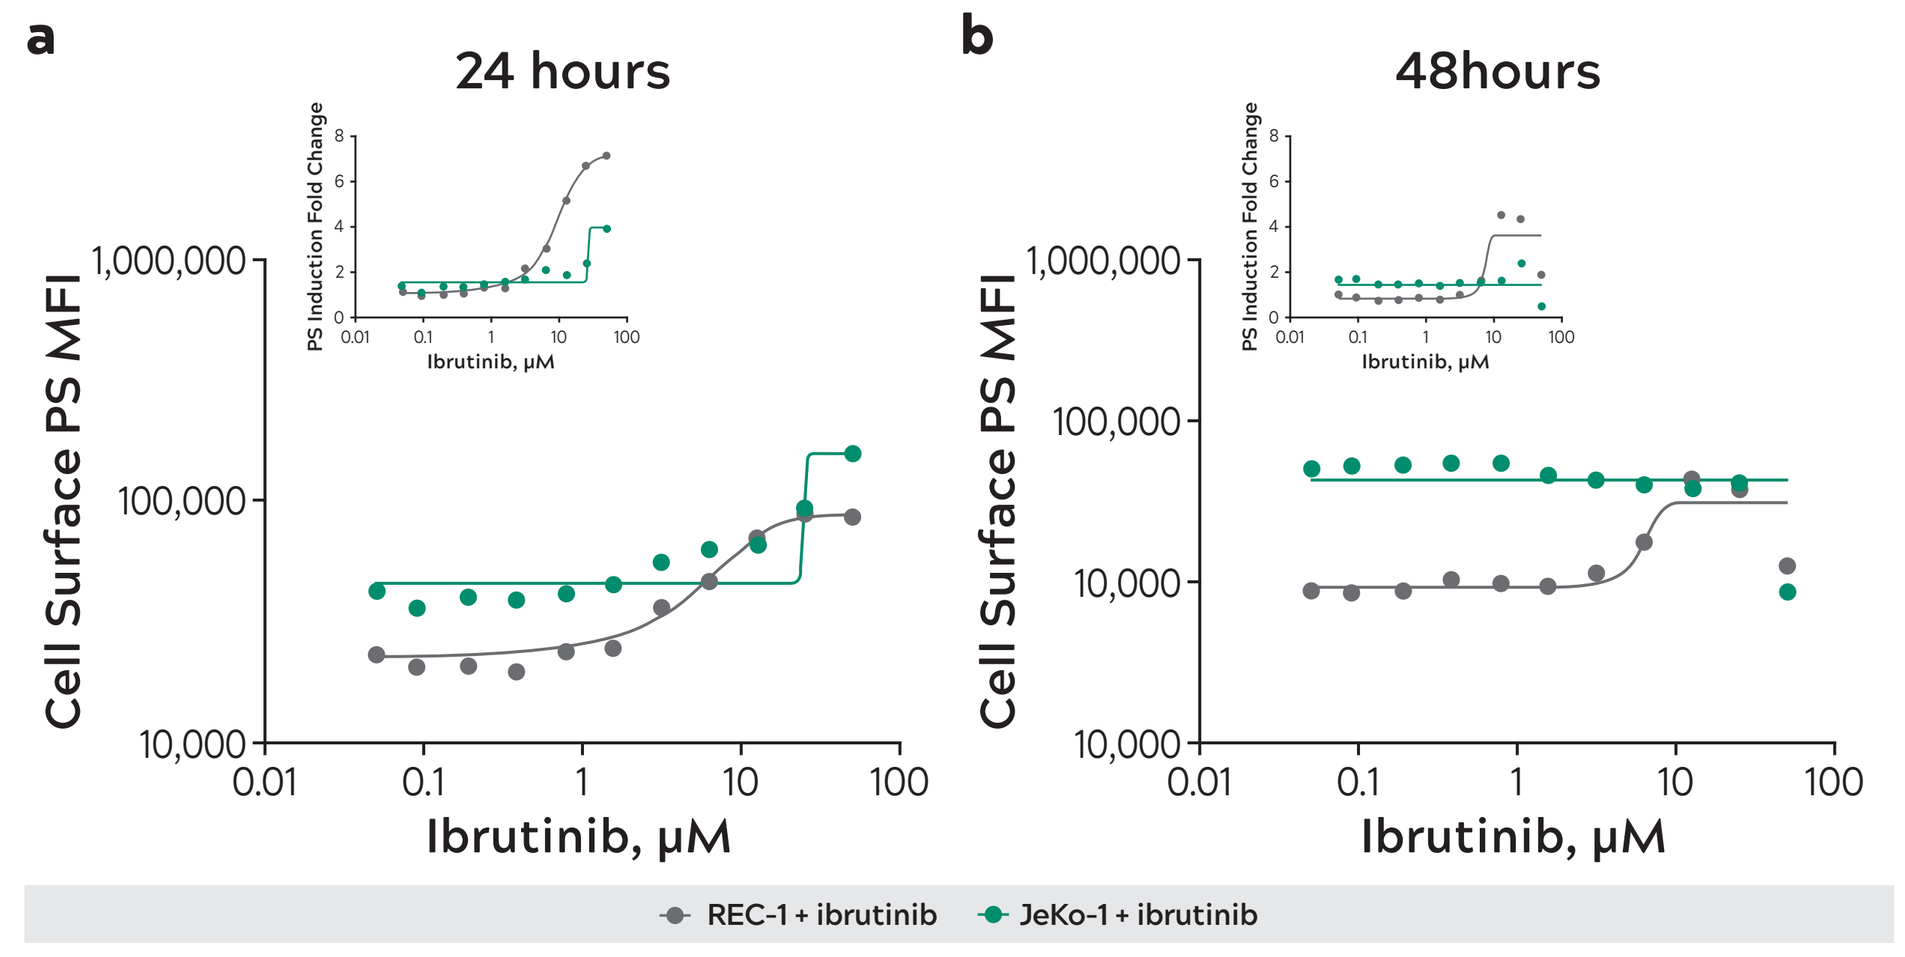
a
24 hours
b
48hours
Cell Surface PS MFI
Ibrutinib, μM
PS Induction Fold Change
REC-1 + ibrutinib
JeKo-1 + ibrutinib

MFI, mean fluorescent intensity; PS, phosphatidylserine.

CER T CELLS DEMONSTRATE PHAGOCYTIC FUNCTION

- CER-1161 and CER-1234 T cells increased engulfment of pHrodo red+ target cells relative to untransduced T cells after 16 and 40 hours of co-culture as measured by FC and fluorescence microscopy (**Figure 4, A to D**)
 - CER constructs containing intracellular signaling domains (ie, CD28, CD3ζ, TLR2) may enhance cytolytic function, leading to enhanced phagocytosis

Figure 4. Phagocytic Activity of REC-1 Cells After Ibrutinib Therapy by CER T Cells.
(**A**) Representative FC histograms showing engulfment of pHrodo red-labeled REC-1 target cells by CER T cells. (**B**) Graphical summary of FC data showing engulfment of pHrodo red-labeled REC-1 target cells by CER T cells. **P*<0.01, ***P*<0.001, ****P*<0.0001; mean + SD, n = 4. (**C**) Representative fluorescence microscopy images (at 40x) showing engulfment of pHrodo red-labeled REC-1 target cells by CER T cells. (**D**) Engulfed pHrodo-green-labeled REC-1 tumor fragments co-localize into Lyso-tracker red-labeled lysosomes.

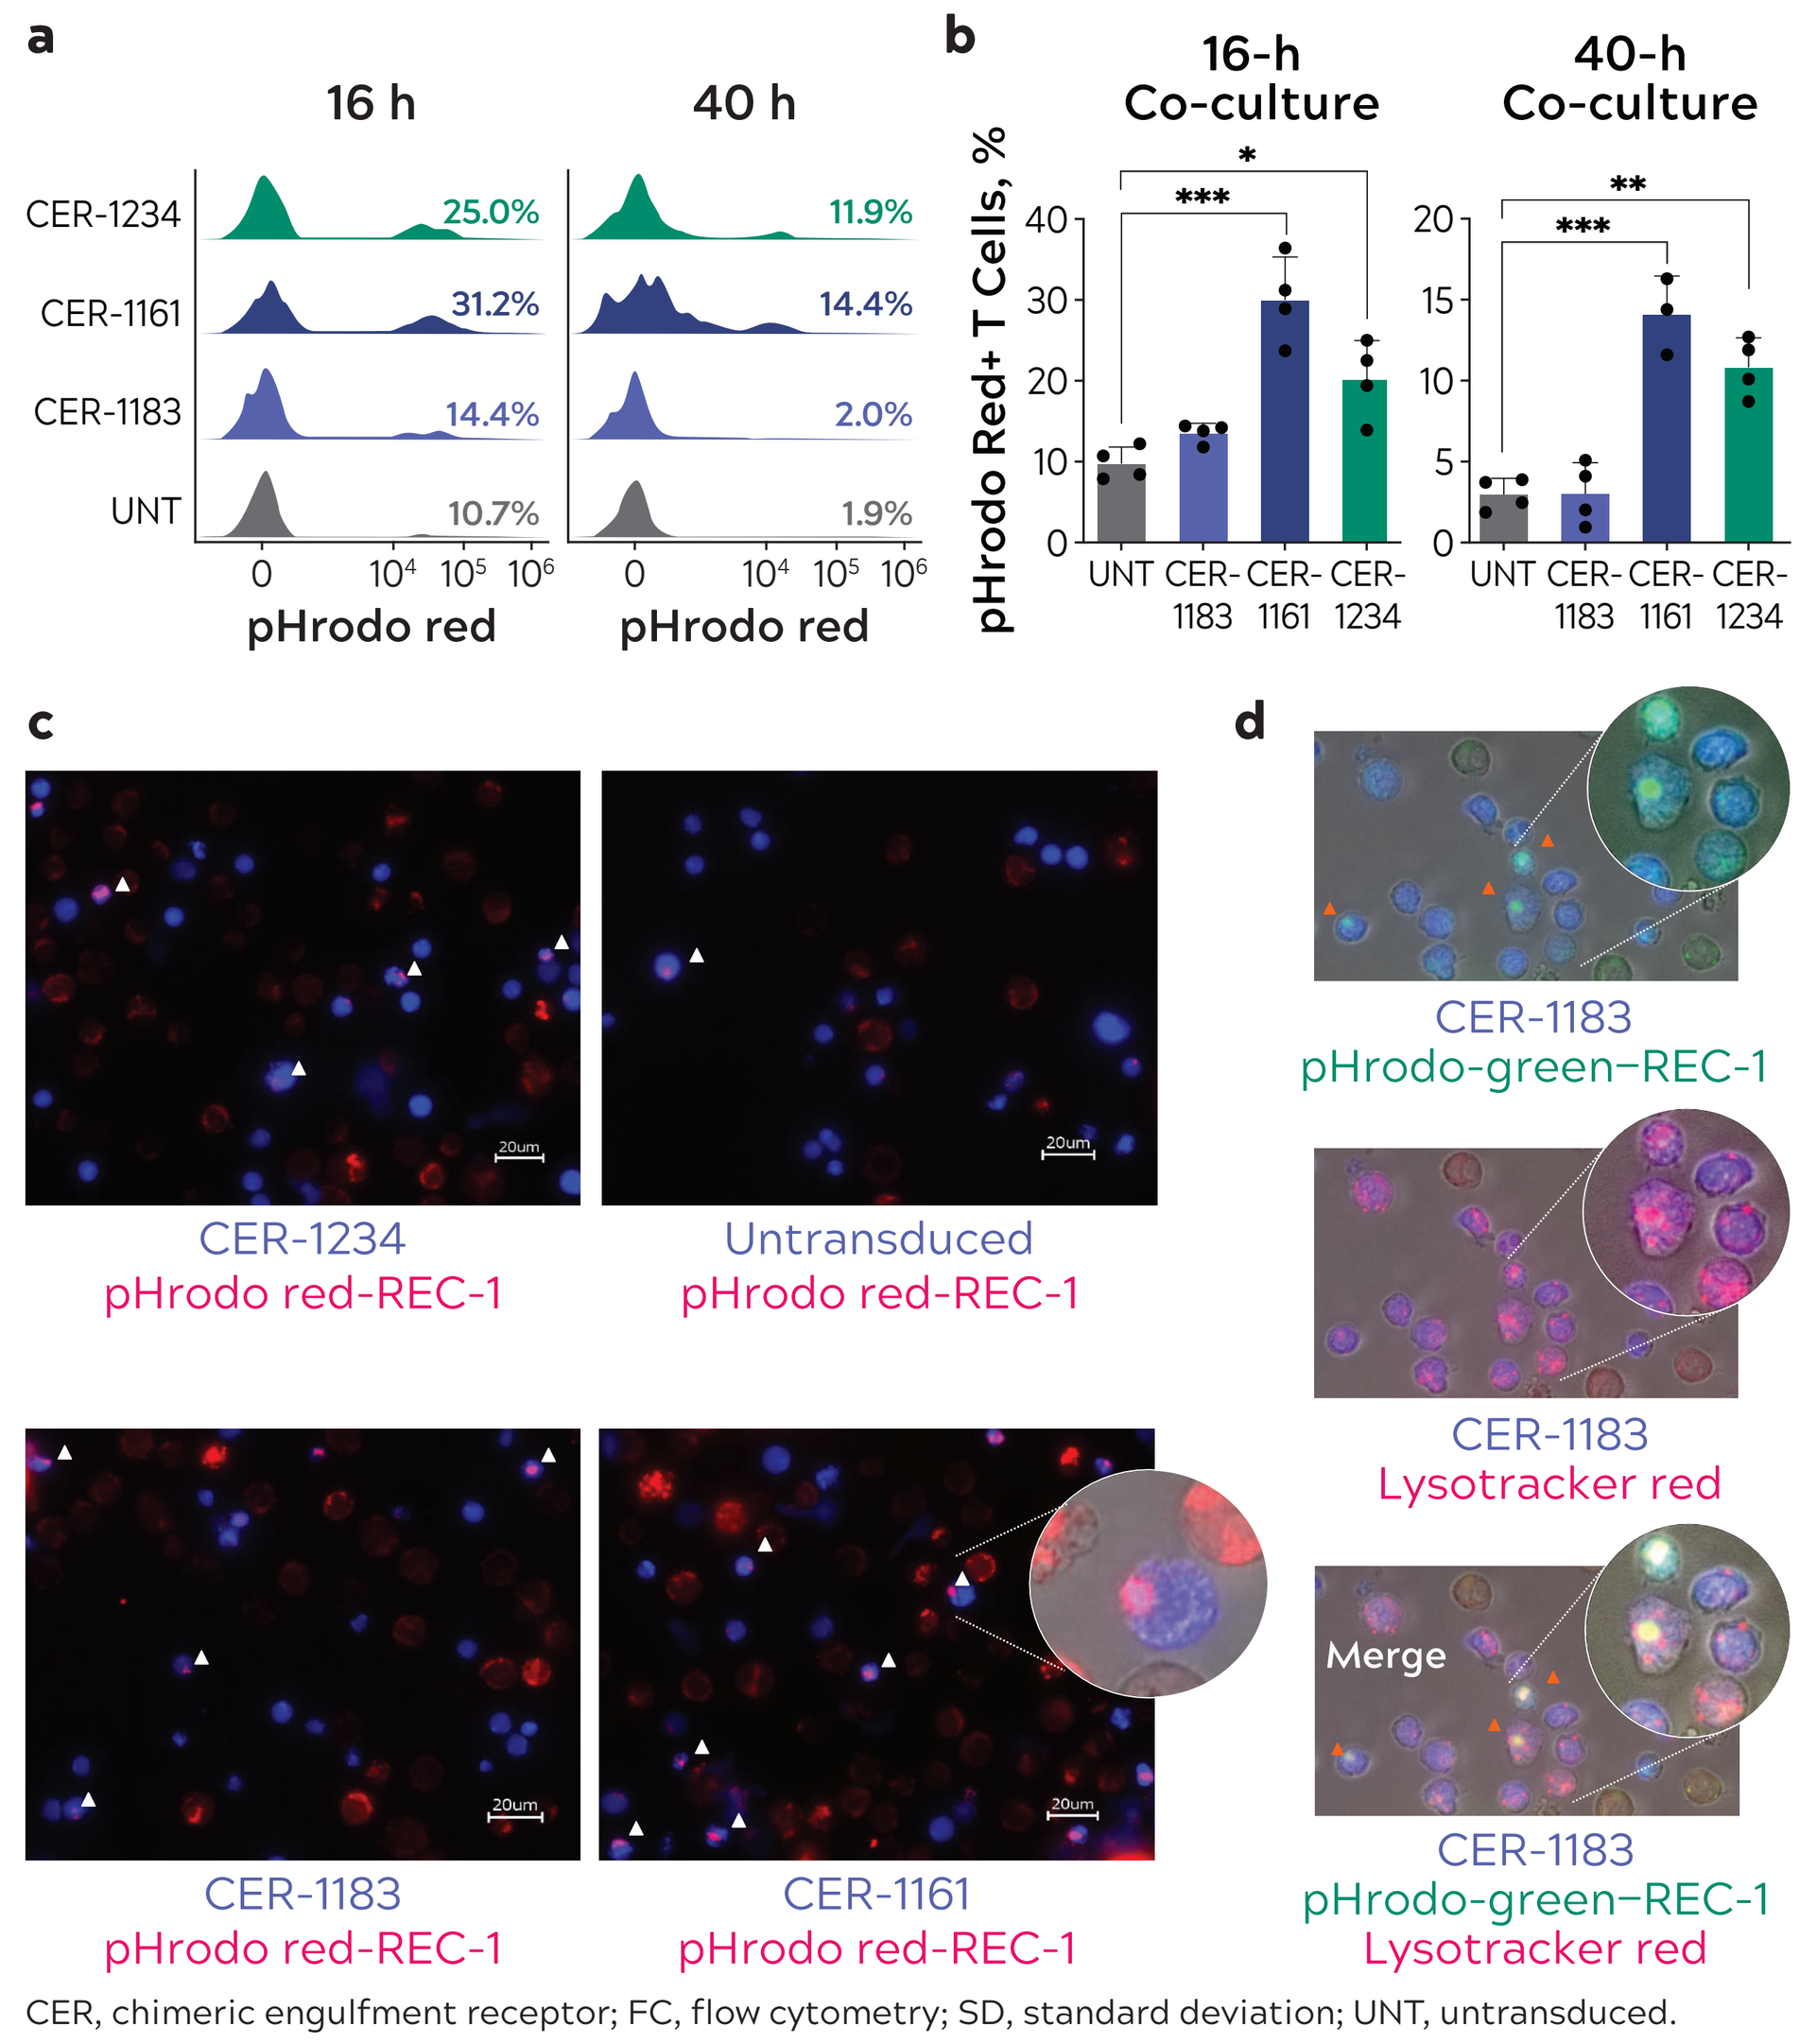
a
16 h
40 h
CER-1234
CER-1161
CER-1183
UNT
25.0%
31.2%
14.4%
10.7%
11.9%
14.4%
2.0%
1.9%
pHrodo red
b
16-h Co-culture
40-h Co-culture
pHrodo Red+ T Cells, %
UNT CER-1183 CER-1161 CER-1234
c
CER-1234 pHrodo red-REC-1
Untransduced pHrodo red-REC-1
CER-1183 pHrodo red-REC-1
CER-1161 pHrodo red-REC-1
d
CER-1183 pHrodo-green–REC-1
CER-1183 Lysotracker red
Merge
CER-1183 pHrodo-green–REC-1 Lysotracker red

CER, chimeric engulfment receptor; FC, flow cytometry; SD, standard deviation; UNT, untransduced.

CER T CELLS DEMONSTRATE CYTOTOXIC ACTIVITY WITH IBRUTINIB

- Tumor cell killing was enhanced by the combination of CER-1234 T cells and ibrutinib (pre-treatment, in co-culture, or both; **Figure 5, A and B**)
 - Co-culture with 0.5 μM ibrutinib reflects the concentration (C_{max}) following standard dosing (560 mg once daily)[3] and had a similar cytotoxic effect as pre-treatment alone or both pre-treatment and co-culture with ibrutinib
 - CER-1183 T cells (no intracellular signaling domain) show no additive effect with ibrutinib
 - CER-1234 T cells alone exhibited limited cytotoxic activity

Figure 5. Cytotoxic Activity of CER T Cells With Ibrutinib in REC-1 Cells.
(**A**) Vehicle pre-treatment followed by +/- 0.5 μM ibrutinib. (**B**) 10 μM ibrutinib pre-treatment followed by +/- 0.5 μM ibrutinib.

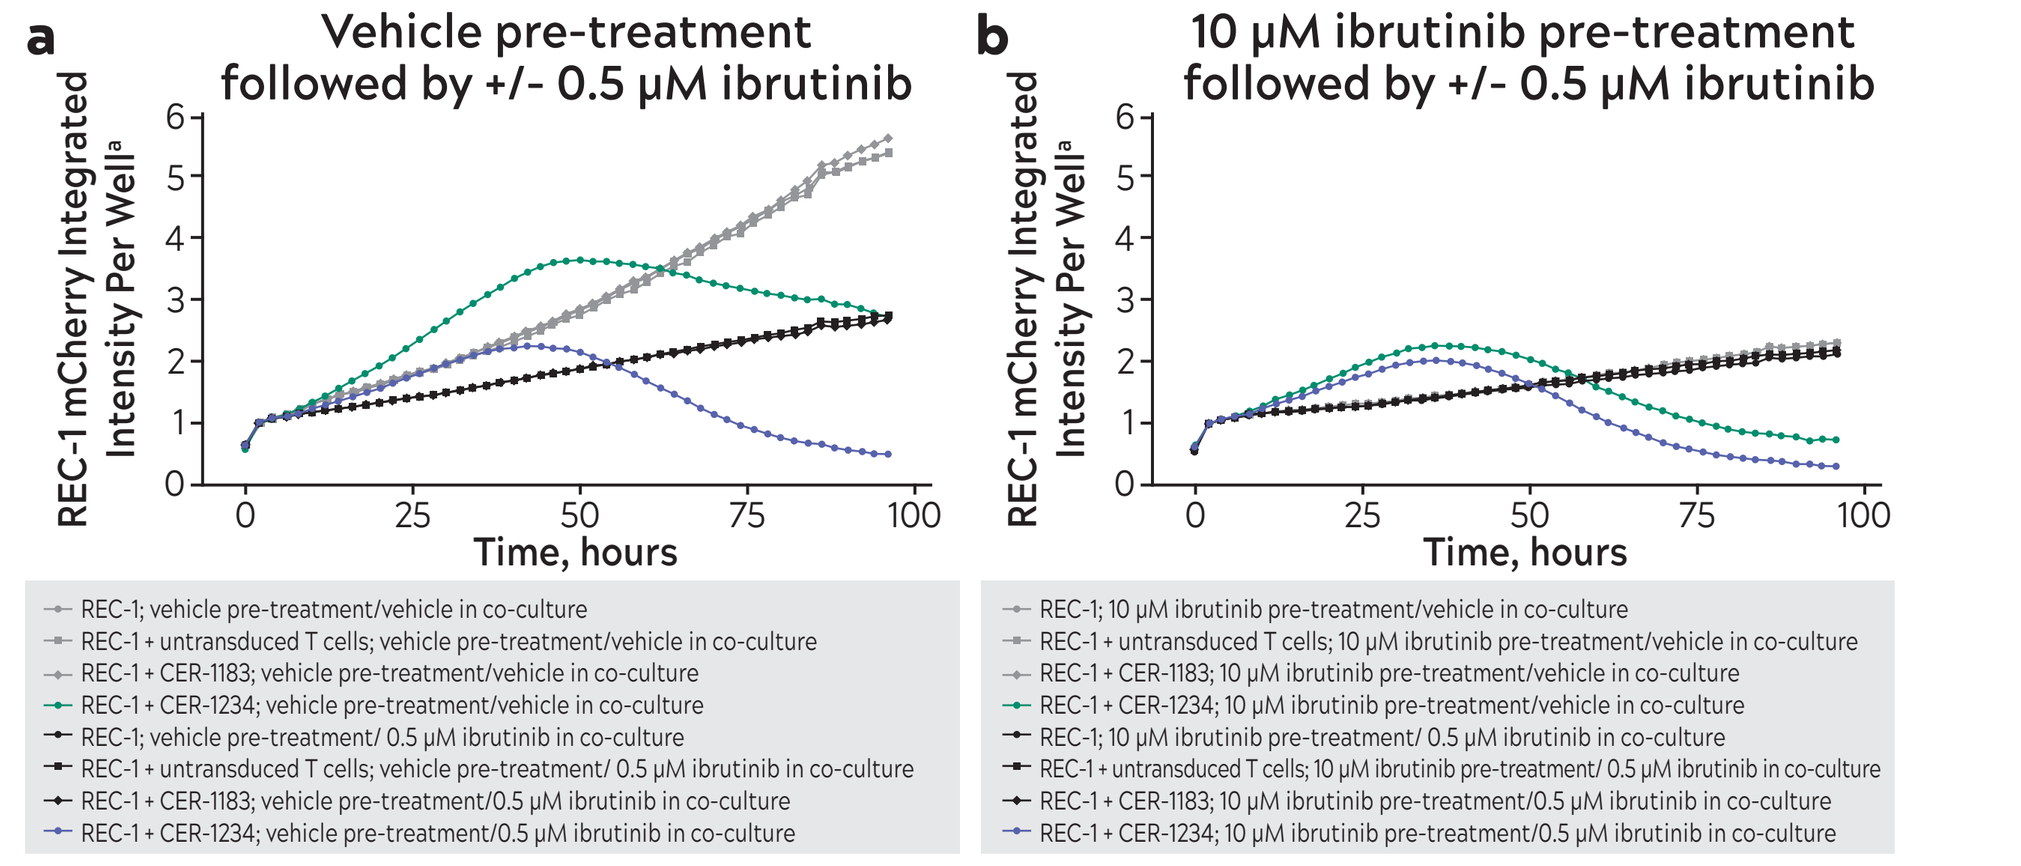
a
Vehicle pre-treatment followed by +/- 0.5 μM ibrutinib
b
10 μM ibrutinib pre-treatment followed by +/- 0.5 μM ibrutinib
REC-1 mCherry Integrated Intensity Per Well[a]
Time, hours
REC-1; vehicle pre-treatment/vehicle in co-culture
REC-1 + untransduced T cells; vehicle pre-treatment/vehicle in co-culture
REC-1 + CER-1183; vehicle pre-treatment/vehicle in co-culture
REC-1 + CER-1234; vehicle pre-treatment/vehicle in co-culture
REC-1; vehicle pre-treatment/ 0.5 μM ibrutinib in co-culture
REC-1 + untransduced T cells; vehicle pre-treatment/ 0.5 μM ibrutinib in co-culture
REC-1 + CER-1183; vehicle pre-treatment/0.5 μM ibrutinib in co-culture
REC-1 + CER-1234; vehicle pre-treatment/0.5 μM ibrutinib in co-culture
REC-1; 10 μM ibrutinib pre-treatment/vehicle in co-culture
REC-1 + untransduced T cells; 10 μM ibrutinib pre-treatment/vehicle in co-culture
REC-1 + CER-1183; 10 μM ibrutinib pre-treatment/vehicle in co-culture
REC-1 + CER-1234; 10 μM ibrutinib pre-treatment/vehicle in co-culture
REC-1; 10 μM ibrutinib pre-treatment/ 0.5 μM ibrutinib in co-culture
REC-1 + untransduced T cells; 10 μM ibrutinib pre-treatment/ 0.5 μM ibrutinib in co-culture
REC-1 + CER-1183; 10 μM ibrutinib pre-treatment/0.5 μM ibrutinib in co-culture
REC-1 + CER-1234; 10 μM ibrutinib pre-treatment/0.5 μM ibrutinib in co-culture

[a]Normalized to 0 days, 2 hours, and 0 minutes.
CER, chimeric engulfment receptor.

CER T CELLS EXPRESS CELL-SURFACE MARKERS OF T-CELL ACTIVATION WHEN CO-CULTURED WITH IBRUTINIB-TREATED REC-1 CELLS

- Co-cultures of CER-1234 T cells and target REC-1 cells in the presence of 0.5 μM ibrutinib increased PD-1 and 4-1BB expression on the T-cell surface (**Figure 6, A and B**)
 - PD-1 and 4-1BB were not increased in co-cultures containing CER-1183 T cells, which lack intracellular signaling domains

Figure 6. Induction of Cell-Surface Markers of T-Cell Activation in Ibrutinib-Treated Co-Cultures of CER T Cells and REC-1 Target Cells.
(**A**) PD-1 expression in co-cultures. (**B**) 4-1BB expression in co-cultures.

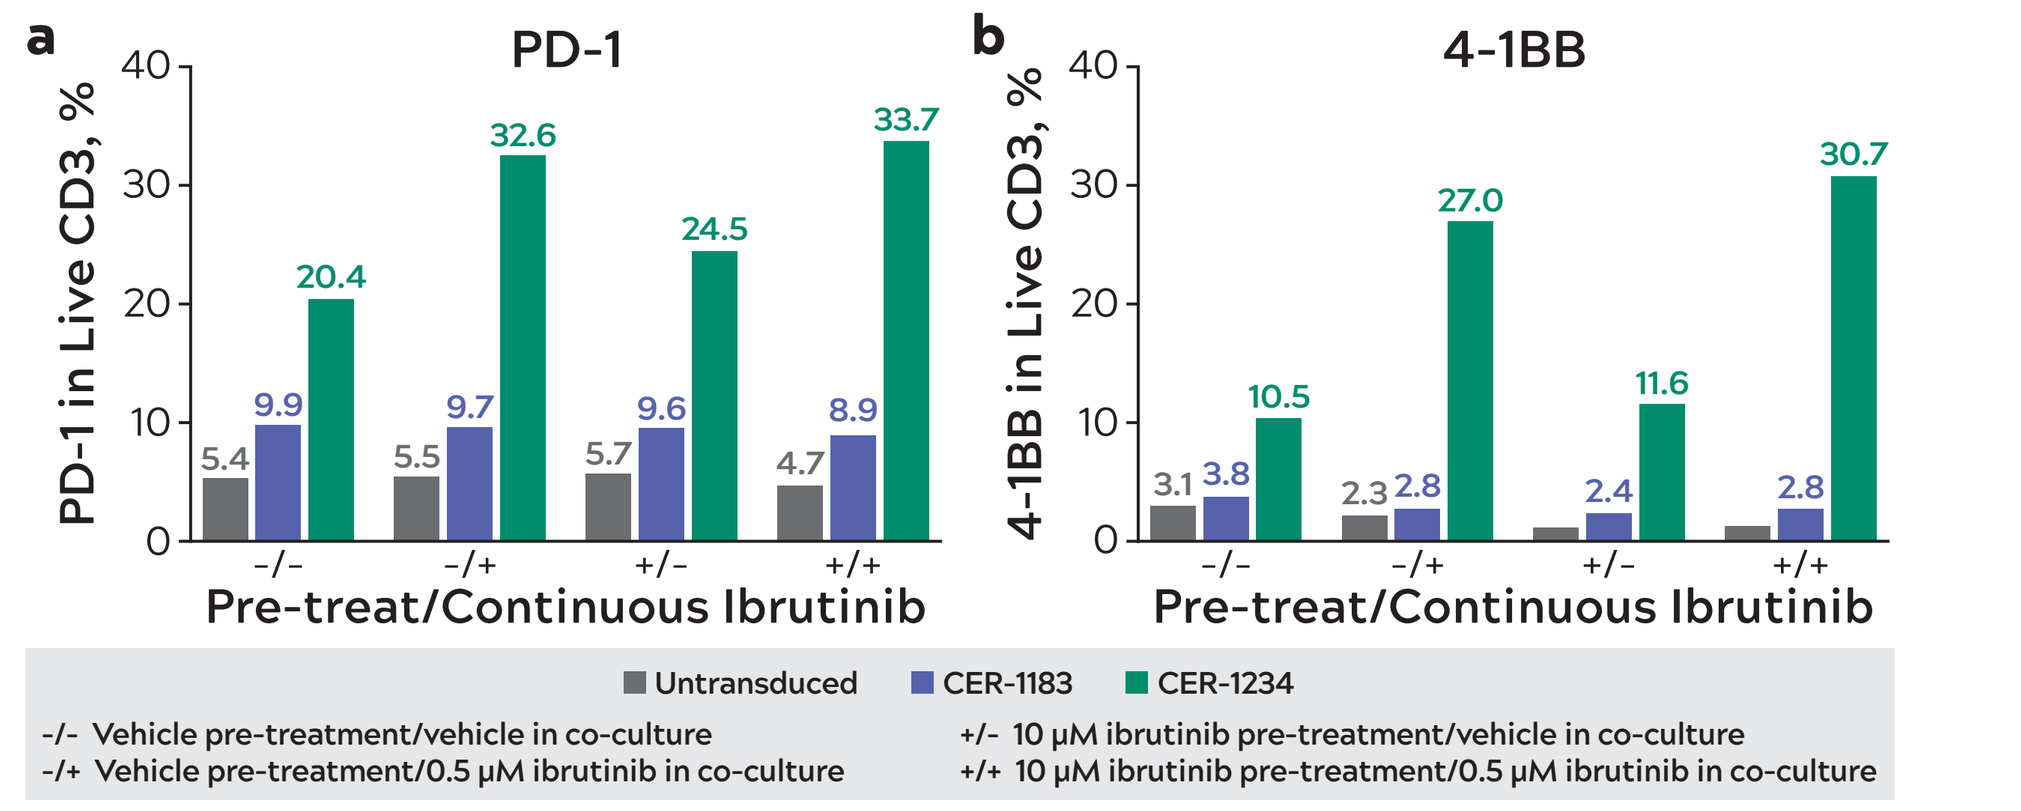
a
PD-1
PD-1 in Live CD3, %
b
4-1BB
4-1BB in Live CD3, %
Pre-treat/Continuous Ibrutinib
-/-
-/+
+/-
+/+
5.4 9.9 20.4
5.5 9.7 32.6
5.7 9.6 24.5
4.7 8.9 33.7
3.1 3.8 10.5
2.3 2.8 27.0
2.4 11.6
2.8 30.7
Untransduced
CER-1183
CER-1234
-/- Vehicle pre-treatment/vehicle in co-culture
-/+ Vehicle pre-treatment/0.5 μM ibrutinib in co-culture
+/- 10 μM ibrutinib pre-treatment/vehicle in co-culture
+/+ 10 μM ibrutinib pre-treatment/0.5 μM ibrutinib in co-culture

CER, chimeric engulfment receptor; PD-1, programmed death-1.

CYTOKINE EXPRESSION WAS INDUCED IN CO-CULTURES OF CER T CELLS AND IBRUTINIB-TREATED REC-1 CELLS

- The following categories of cytokines were induced in co-cultures of CER-1234 T cells and target REC-1 cells in the presence of 0.5 μM ibrutinib (**Figure 7, A to E**)
 - T1: tumor necrosis factor alpha (TNF-α), interferon gamma (IFN-γ), and interleukin (IL)-10
 - T2: IL-5
 - Effector: granzyme B
- Cytokine expression was not induced in co-cultures containing CER-1183 T cells, which contain Tim-4 but lack intracellular signaling domains
- IL-2 (homeostatic), IL-4 (T2), and IL-6 (inflammatory) were nominally increased
- IL-12p70 (APC), IL-17A (inflammatory), and IL-1β (inflammatory) were at the limit of detection for all co-culture conditions

Figure 7. Induction of Cytokines in Ibrutinib-Treated Co-Cultures of CER T cells and REC-1 Target Cells.
(**A**) TNFα induction. (**B**) IFNγ. (**C**) IL-5 induction. (**D**) IL-10 induction. (**E**) granzyme B induction.

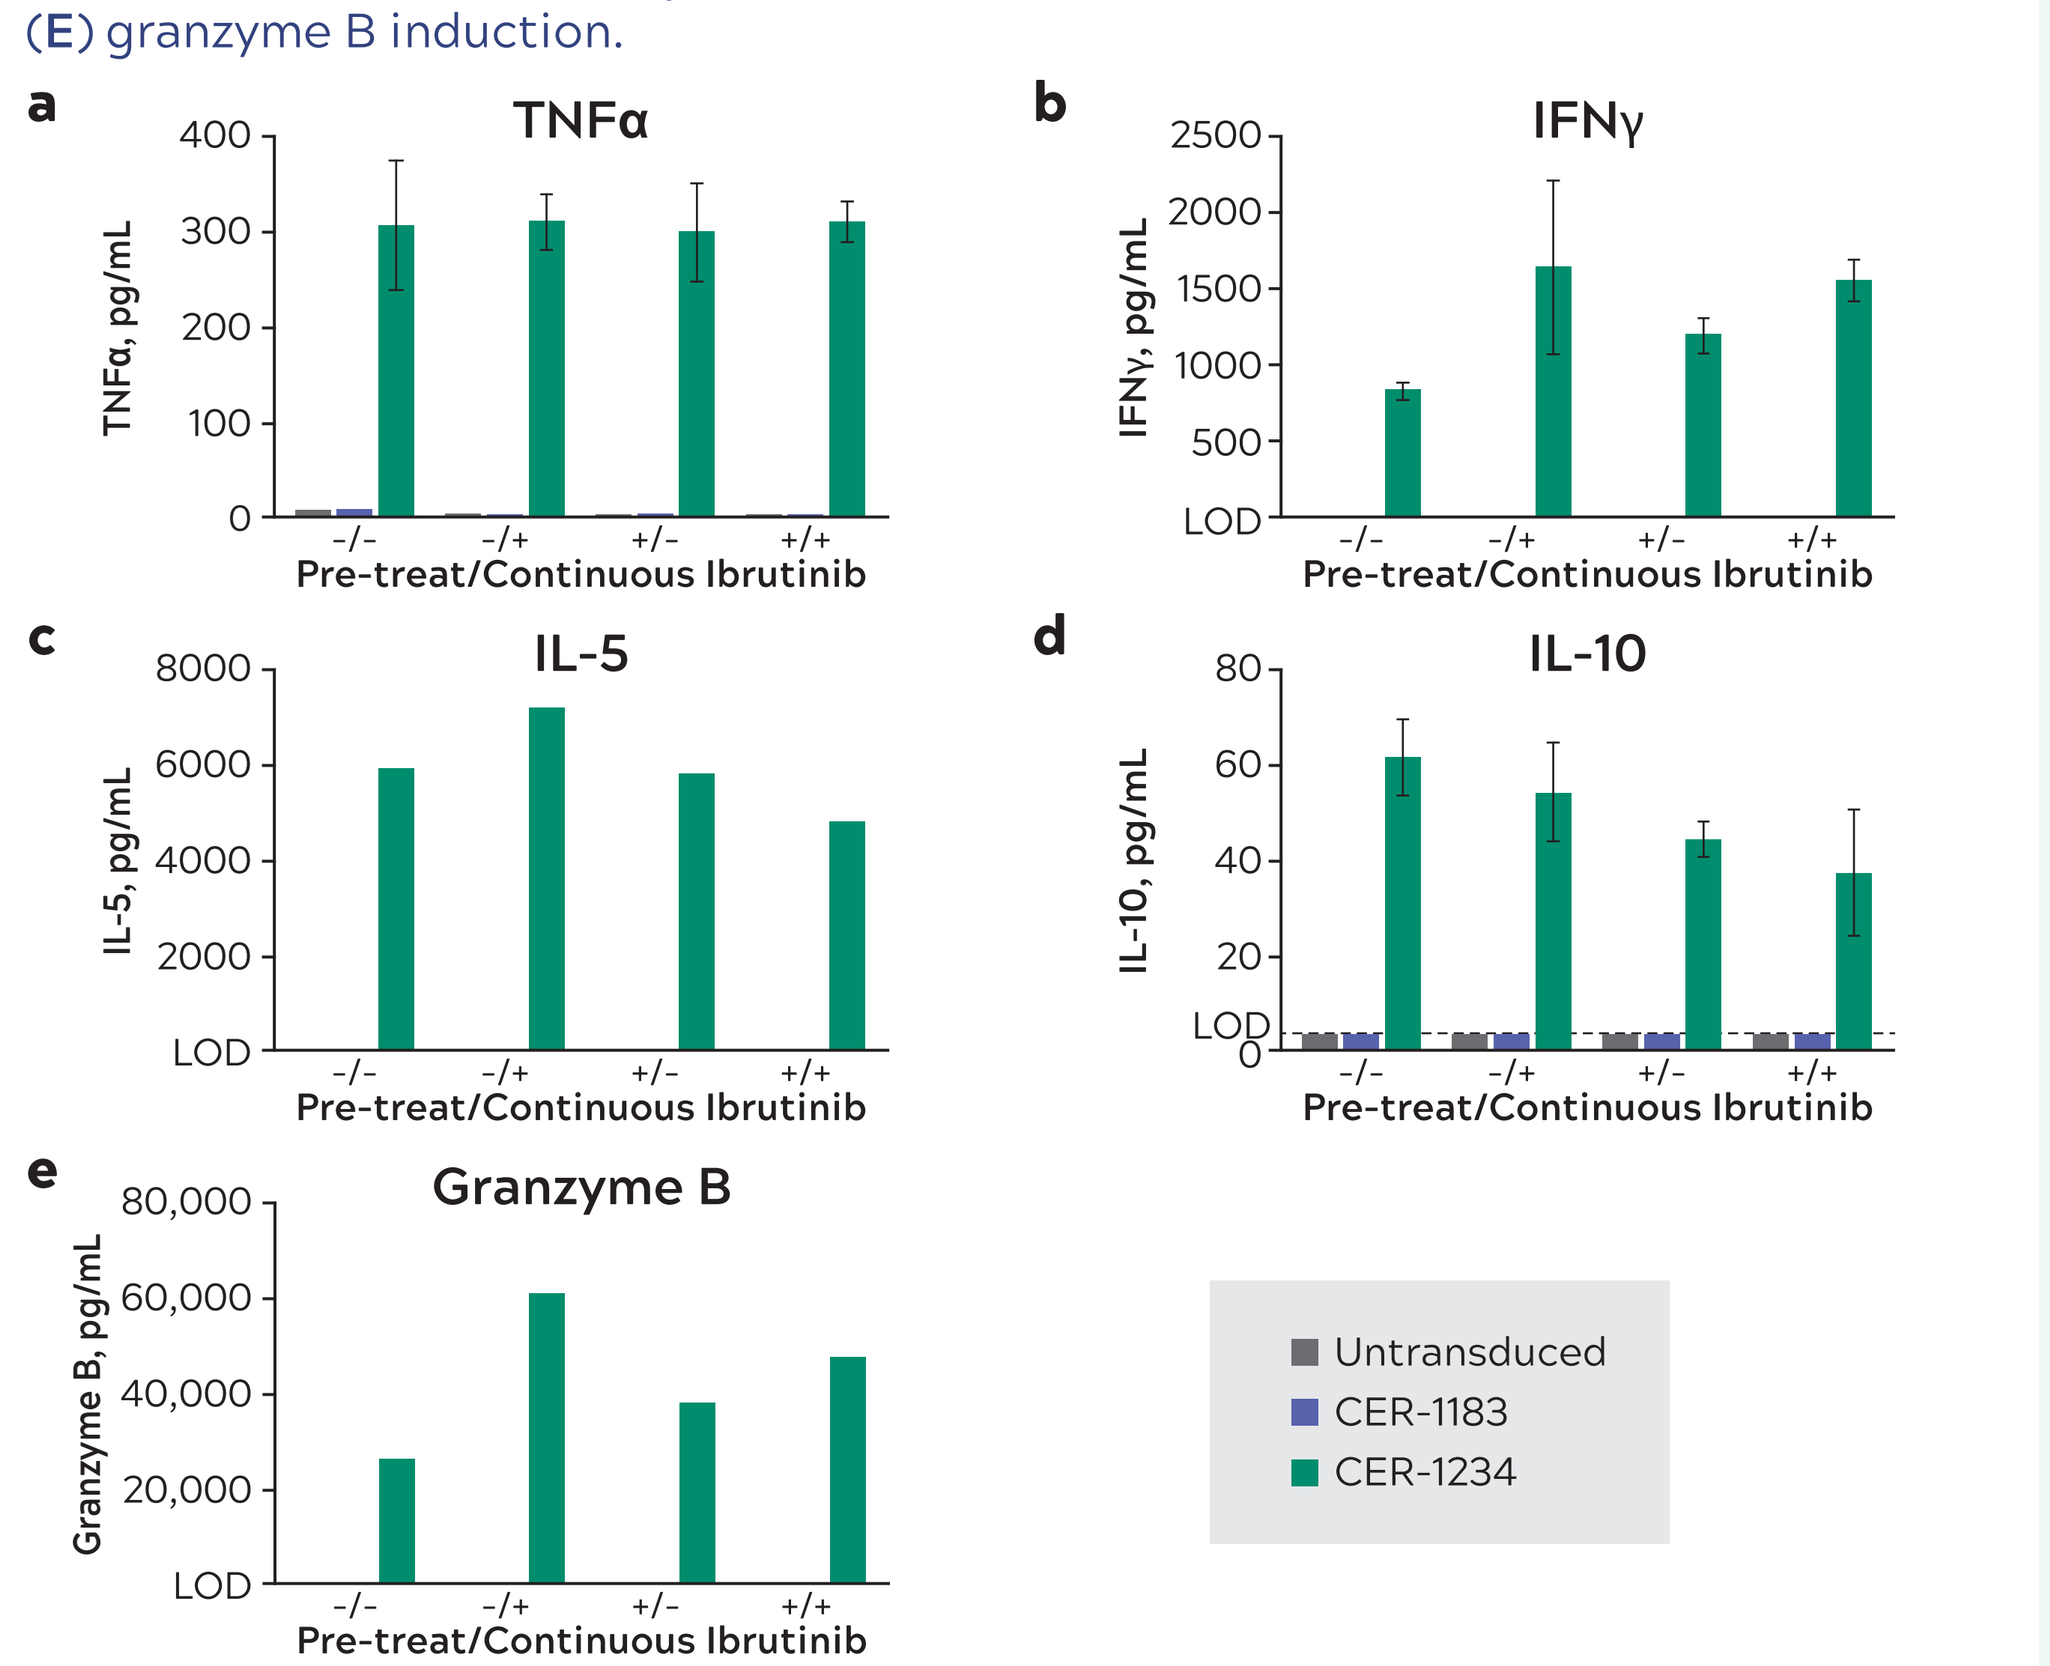
a
TNFα
TNFα, pg/mL
b
IFNγ
IFNγ, pg/mL
c
IL-5
IL-5, pg/mL
d
IL-10
IL-10, pg/mL
e
Granzyme B
Granzyme B, pg/mL
LOD
Pre-treat/Continuous Ibrutinib
-/-
-/+
+/-
+/+
Untransduced
CER-1183
CER-1234

CER, chimeric engulfment receptor; IFNγ, interferon gamma; IL-2, interleukin-2; IL-4, interleukin-4; IL-5, interleukin-5; IL-6, interleukin-6; IL-10, interleukin-10; LOD, limit of detection; TNFα, tumor necrosis factor alpha.

CER T CELLS CONTAINING A TLR2 DOMAIN DEMONSTRATE ENHANCED ANTIGEN-PRESENTING CELL-LIKE FUNCTION IN VITRO

- In an E7 HLA-A2 class I-restricted TCR system (**Figure 2**), the addition of the TLR2 intracellular signaling domain in CER-1107 T cells increased proliferation of autologous E7 TCR T cells relative to that observed for CER-247 T cells, indicative of enhanced E7 antigen presentation by CER-1107 T cells pulsed with E7 peptides (**Figure 8, A and B**)
- CER T cell-induced proliferation is HLA-dependent, confirming the role of antigen presentation in the CER-mediated activity (**Figure 8C**)

Figure 8. Antigen-Presenting Cell-Like Function of CER T Cells In Vitro.
(**A**) Histograms of FC data show proliferation of E7 TCR T cells marked by CTV dilution. (**B**) Quantitation of proliferated E7 TCR T cells. (**C**) Percentage of proliferated E7 TCR cells in culture among live CD3+ cells determined by FC based on staining of CTV low E7 TCRb+ cells.

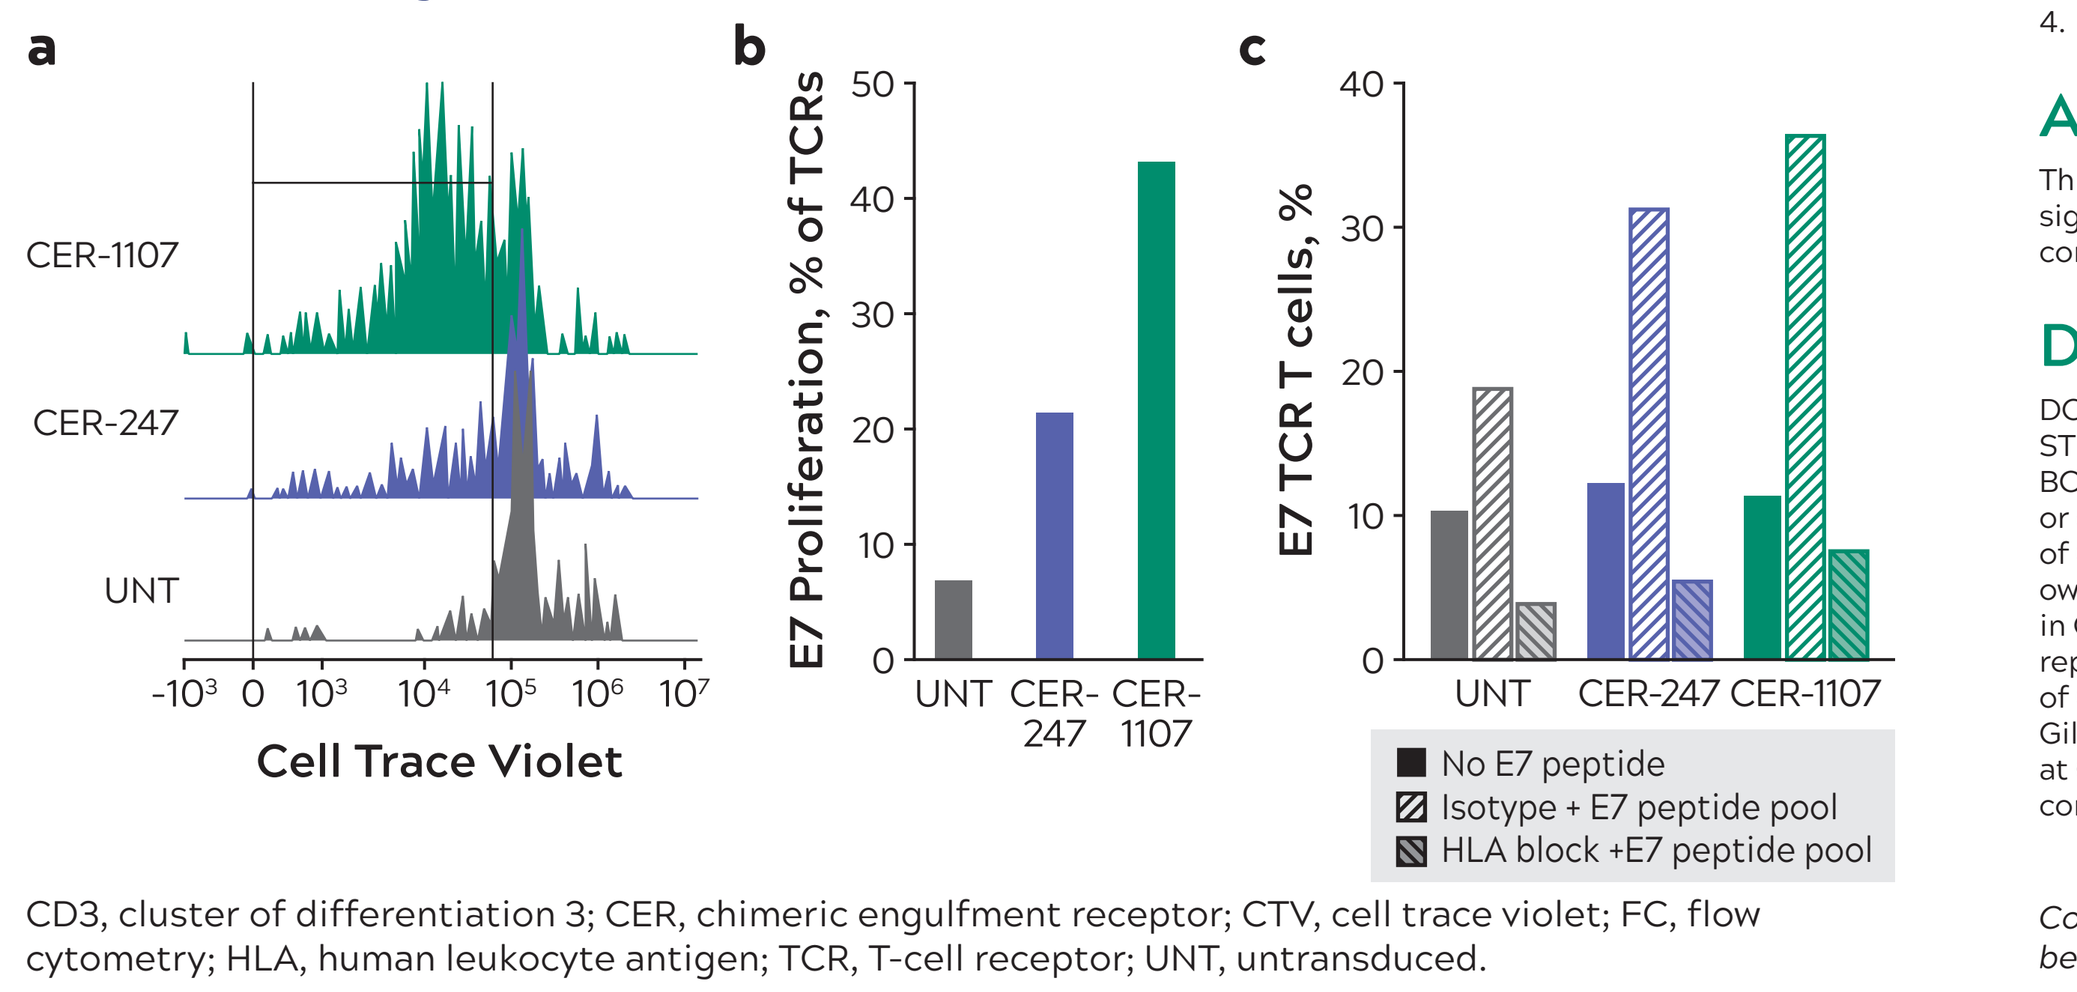
a
CER-1107
CER-247
UNT
Cell Trace Violet
b
E7 Proliferation, % of TCRs
UNT CER-247 CER-1107
c
E7 TCR T cells, %
UNT CER-247 CER-1107
No E7 peptide
Isotype + E7 peptide pool
HLA block +E7 peptide pool

CD3, cluster of differentiation 3; CER, chimeric engulfment receptor; CTV, cell trace violet; FC, flow cytometry; HLA, human leukocyte antigen; TCR, T-cell receptor; UNT, untransduced.

CONCLUSIONS

- Novel CER T cells containing a Tim-4 extracellular domain and either CD28 and CD3ζ or CD28, CD3ζ, and TLR2 intracellular signaling domains can kill MCL tumor cells, capture tumor cell fragments, and induce expression of T-cell activation markers and cytokines in vitro
- The TLR2 domain enhanced the ability of CER T cells to present an exogenous soluble antigen and activate MHC class I-restricted T cells, a function previously demonstrated to be a barrier to effective antigen presentation in T cells[1]
- T-cell antigen capture and presentation capabilities, combined with inducible and target-specific cytotoxic function in single T cells, suggest the potential for secondary immune responses via activation and enhancement of endogenous anti-tumor immunity (**Figure 9**)
- CER T cells offer the potential for combination with multiple targeted therapies and application to both hematologic and solid tumors
- CER T cells may also provide clinical benefit as a single therapeutic agent due to the well documented instability of malignant cell membrane symmetry related to increased proliferative capacity and/or alterations in key pathways involved in cell membrane homeostasis[4-7]

Figure 9. Proposed Mechanism of Action of CER T Cells.
CER T cells are designed to combine adaptive T-cell cytotoxicity with innate functions of phagocytosis and antigen processing and presentation, leading to secondary immune responses.

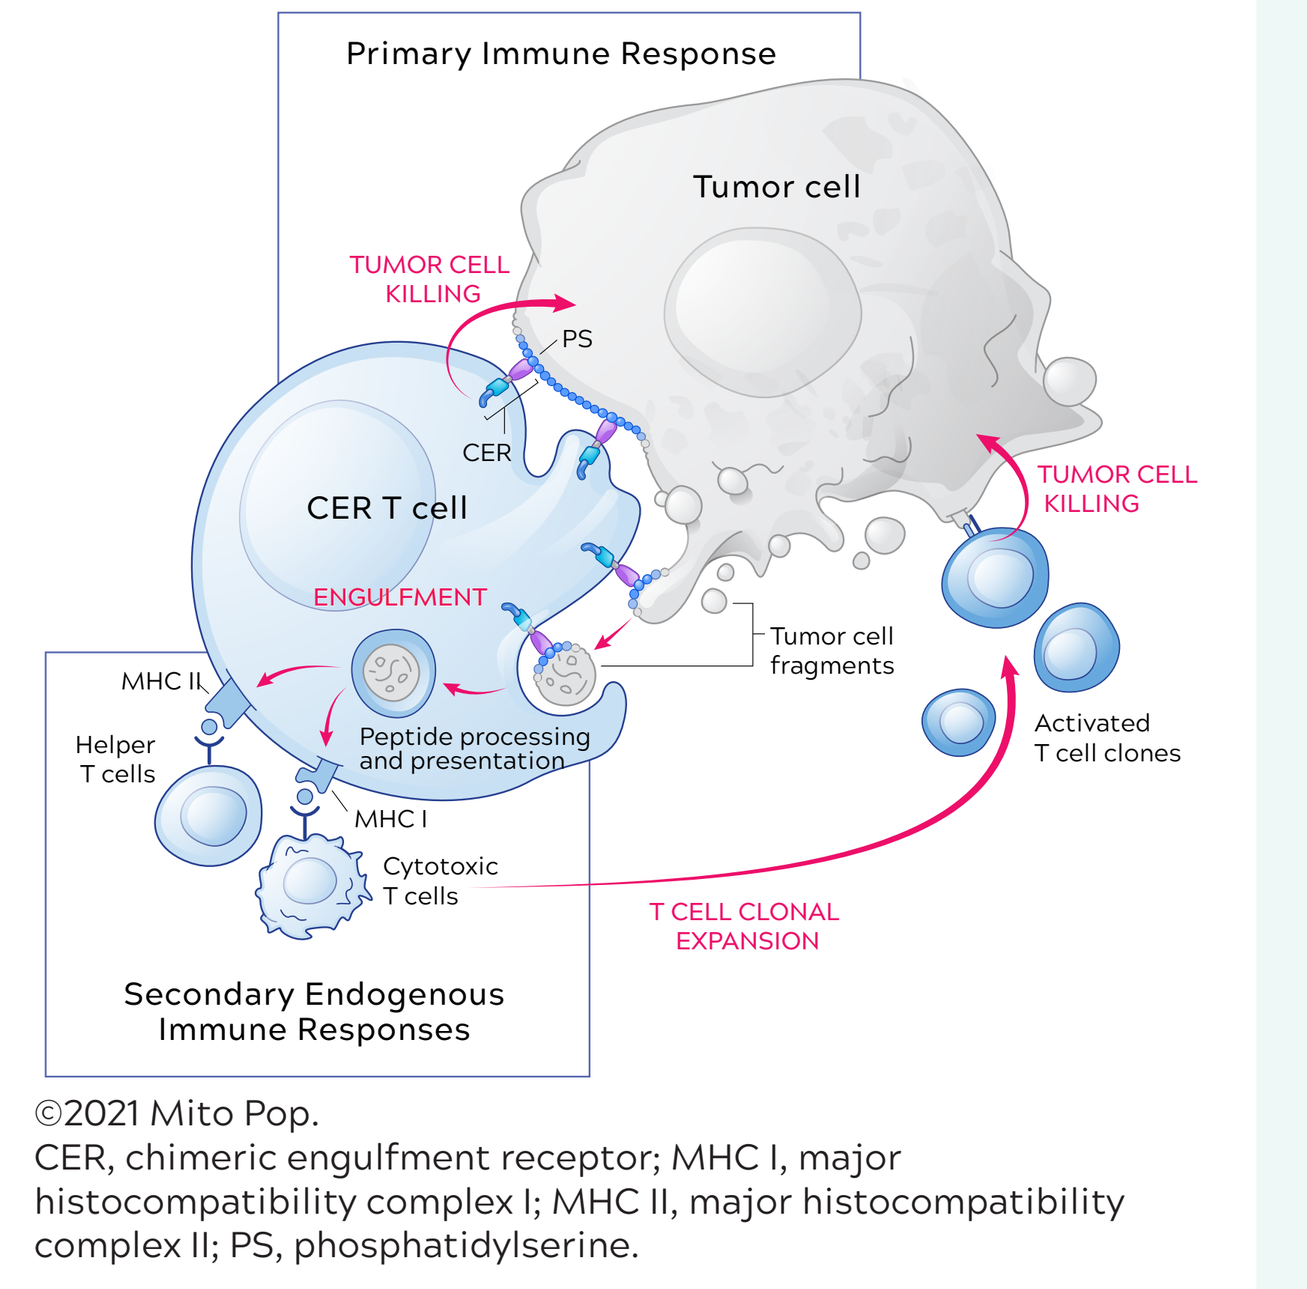
Primary Immune Response
Tumor cell
TUMOR CELL KILLING
PS
CER
CER T cell
ENGULFMENT
Tumor cell fragments
TUMOR CELL KILLING
Activated T cell clones
MHC II
Helper T cells
Peptide processing and presentation
MHC I
Cytotoxic T cells
T CELL CLONAL EXPANSION
Secondary Endogenous Immune Responses

©2021 Mito Pop.
CER, chimeric engulfment receptor; MHC I, major histocompatibility complex I; MHC II, major histocompatibility complex II; PS, phosphatidylserine.

REFERENCES

1. Lanzavecchia A, et al. *Nature*. 1988;334:530.
2. Caronni N, et al. *Nat Commun*. 2021;12:2237.
3. Advani RH, et al. *J Clin Oncol*. 2013;31:88.
4. Riedl S, et al. *Biochim Biophys Acta*. 2011;1808:2638.
5. Birge RB, et al. *Cell Death Differ*. 2016;23:962.
6. Ennishi D, et al. *Nat Med*. 2020;26:577.
7. Vallabhapurapu AD, et al. *Oncotarget*. 2015;6:34375.

ACKNOWLEDGMENTS

This study was sponsored by CERo Therapeutics. Maggie Bobbin, Ankit Bhatta, and Vishal Sharma, from CERo Therapeutics, contributed significantly to this work. Editorial support was provided by Juan Sanchez-Cortes, PhD, from Nexus Global Group Science, a Vaniam Group company, and funded by CERo Therapeutics.

DISCLOSURES

DC is an employee of CERo Therapeutics, reports stocks or other ownership in CERo Therapeutics, and holds patents with CERo Therapeutics. ST is an employee of CERo Therapeutics, reports stocks or other ownership in CERo Therapeutics, and holds patents with CERo Therapeutics. BC is an employee of CERo Therapeutics and holds patents with CERo Therapeutics. LN is an employee of CERo Therapeutics, reports stock or other ownership in Assembly Biosciences, and holds patents with Assembly Biosciences. JC is an employee of CERo Therapeutics and has been an employee of Kite Pharma and Gilead Sciences. He reports stock or other ownership in Gilead Sciences. JB is an employee of CERo Therapeutics and reports stocks or other ownership in CERo Therapeutics. RV is an employee of CERo Therapeutics and has been an employee of Kite Pharma. He reports stock or other ownership in CERo Therapeutics, Gilead Sciences, AbbVie, and Merck. JMR is an employee of CERo Therapeutics and has been an employee of Gilead Sciences. He reports stock or other ownership in Gilead Sciences and CERo Therapeutics, has received consulting and advisory fees from Amunix, and holds patents at Gilead Sciences. LC reports stock or other ownership at CERo Therapeutics, and has received honoraria and consulting fees from CERo Therapeutics. KD and LJ have no disclosures.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Poster presented at the Society for Immunotherapy of Cancer (SITC) 36th Annual Meeting, November 10-14, 2021.