TIM-4-L-targeting CER-1236 Engineered T cells Eliminate Adverse-Risk AML Xenografts and Exhibit Favorable Manufacturing Product Attributes for Clinical Translation

Brandon Cieniewicz[1], Ankit Bhatta[1], Edson Oliveira[1], Zhuo Yang[1], Bi Yu[1], Hongxiu Ning[1], Daniel Corey[1]

[1] CERo Therapeutics Holdings, Inc. South San Francisco, CA 94080

Corresponding Author: Daniel Corey, dcorey@cero.bio

INTRODUCTION

Engineered T cells targeted to malignant cells have dramatically changed patient outcomes in B cell malignancies, leading to cases of long-term cures in B cell malignancies. Despite this success in B cell malignancies, it has proven difficult to adapt engineered T cell therapy to other indications that frequently lack cell surface proteins specific to cancer cells.

Here we describe a Chimeric Engulfment Receptor (CER) T cell, engineered with a natural TIM-4 binding domain that recognizes the structural phospholipid Phosphatidylserine (PS), or TIM-4-L. TIM-4-L is sequestered to the inner leaflet of the plasma membrane in healthy cells, but can be exposed as an eat-me signal during apoptosis to mark cells for phagocytosis and prevent inflammation. Interestingly, TIM-4-L has been shown to be aberrantly upregulated on multiple cancer subtypes, including AML, where it can play a role in immune evasion. In order to better understand the prevalence of TIM-4-L exposure on malignant cells, we evaluated a panel of primary samples from patients with hematological malignancies to quantify their cell surface TIM-4-L. Additionally, we tested the responses of CER-1236, a CER T cell developed to mediate potent effector function and a novel phagocytic function, against representative AML cancer cell lines, including canonical effector functions such as cytotoxicity, proliferation, and cytokine secretion, as well as CER-specific functions, such as engulfment of TIM-4-L+ targets and cross-presentation of processed antigen.

RESULTS

We screened primary, treatment-naïve PBMC samples from AML, MCL, and CLL patients, as well as AML bone marrow aspirates for baseline positivity for PS by flow cytometry. Malignant cells from tested indications had higher TIM-4-L exposure than healthy control cells. In addition to higher frequency of TIM-4-L positive cells, TIM-4-L per cell expression was also higher in malignant cells.

We developed a clinic-ready production protocol for CER-1236 T cells using the CliniMACS Prodigy and determined that CER-1236 T cells had high transduction efficiency with an acceptable VCN. CER-1236 T cells had a favorable memory phenotype favoring CCR7+ cell subsets, and responded to TIM-4-L in a dose dependent manner. CER-1236 T cells also showed the novel ability to phagocytose TIM-4-L+ targets, and subsequently cross-present antigen to bystander T cells in a TLR2- and HLA class I-dependent manner.

We tested the function of CER-1236 T cells against the Kasumi-1 p53mut or MV-4-11 FLT3-ITD AML cell lines and observed potent cytotoxicity as well as induction of T cell proliferation, and IFN-γ secretion after co-culture at E:T ratios as low as 1:1. We next tested the function of CER-1236 T cells in vivo. Murine TIM-4-L is chemically identical to human TIM-4-L, making mice a relevant safety model as well. Here CER-1236 showed potent cytotoxic function against Kasumi-1 AML in vivo, leading to tumor elimination. CER-1236 T cells showed early expansion, but contracted after tumor elimination without indication of on-target off-tumor toxicity. We determined that healthy murine HSPCs had negligible cell surface TIM-4-L, and the presence of highly engrafted tumor did not lead to TIM-4-L induction on healthy bystander bone marrow cells.

Lastly, we determined that factors secreted by CER-1236 T cells after antigen stimulation could enhance cell surface TIM-4-L on both Kasumi-1 and MV-4-11 AML cells.

CONCLUSIONS

Aberrant TIM-4-L expression is an underappreciated and difficult to capture feature of malignant cells across multiple indications. Healthy cells, however, show tight regulation of cell surface TIM-4-L. TIM-4-L specific CER-1236 T cells can mediate potent anti-tumor effects in vitro and in vivo without harm to healthy cells, as well as mediate unique antigen presentation capacity and secrete factors that can further potentiate TIM-4-L exposure.

CER-1236 Receptor

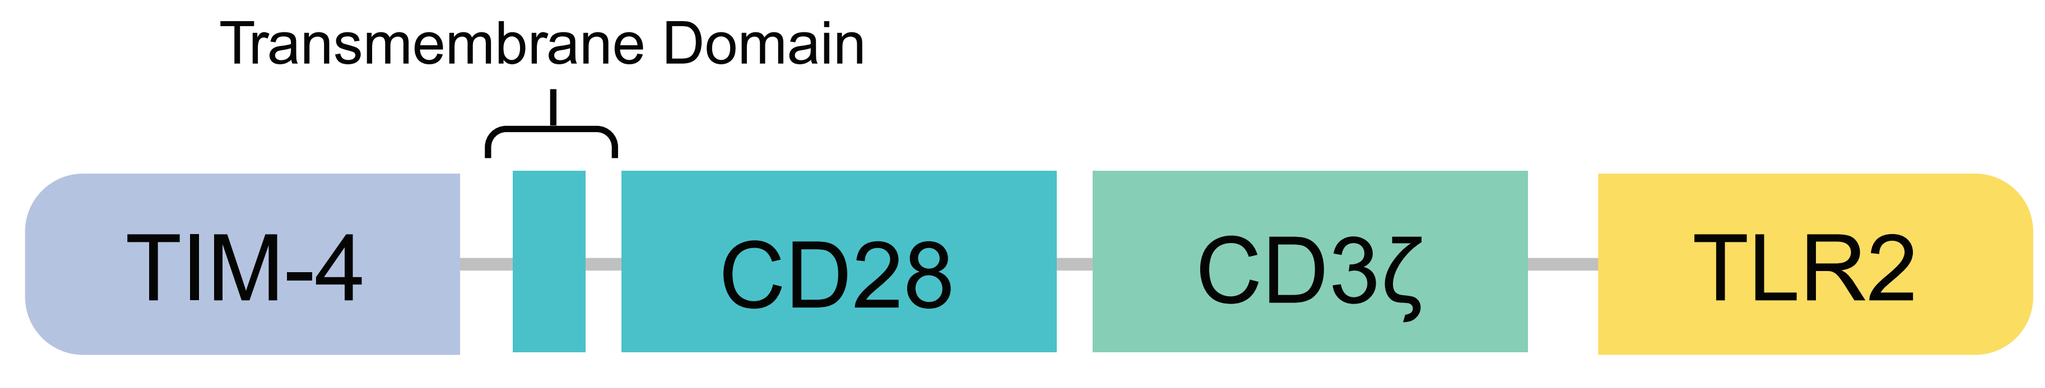
Transmembrane Domain
TIM-4
CD28
CD3ζ
TLR2

Extracellular Domain
- Specific binding to PS and not other phospholipids
- Intrinsic phagocytic function

Canonical T cell signaling domains
- Mediate antigen-specific T cell activation, anti-tumor cytotoxicity, proliferation, and production of effector cytokines
- CD28 co-stimulatory domain mediates rapid and potent T cell activation

Innate signaling domain
- Augments phagocytic and cross-presentation functions
- Supports T cell effector functions

RESULTS

FIGURE 1 Elevated cell surface PS observed on treatment-naive hematologic tumors

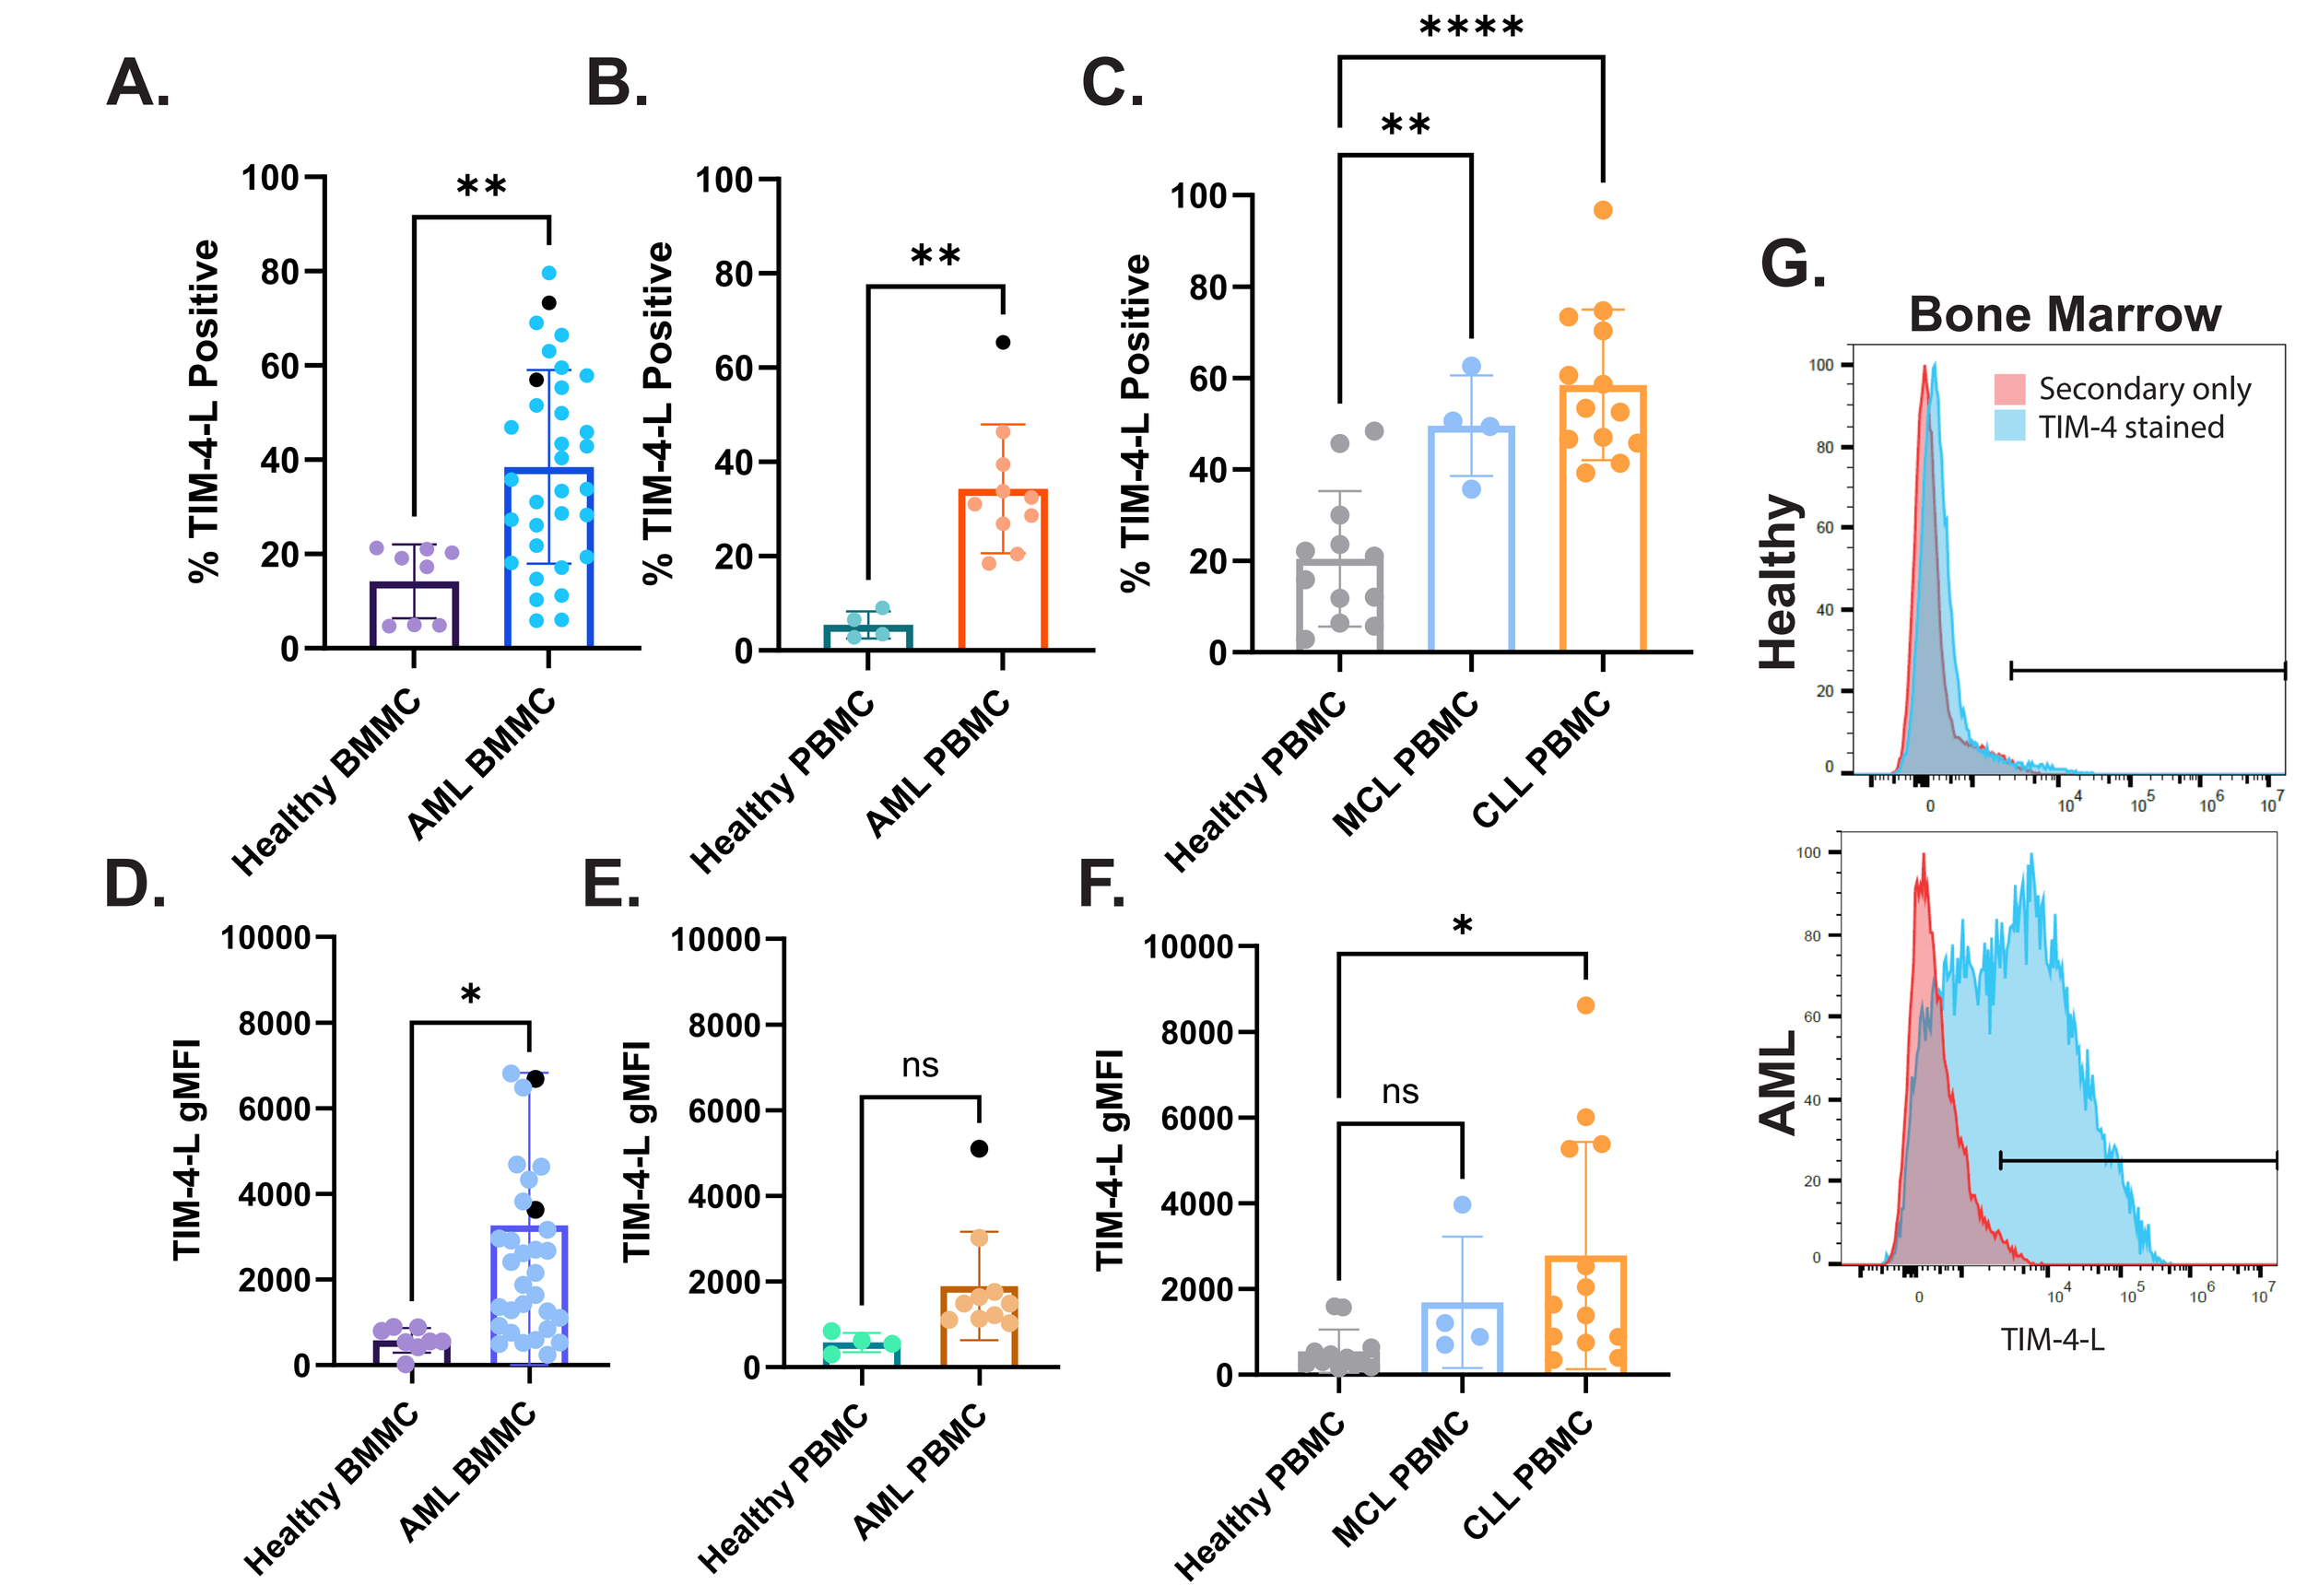
A.
% TIM-4-L Positive
Healthy BMMC
AML BMMC
B.
Healthy PBMC
AML PBMC
C.
Healthy PBMC
MCL PBMC
CLL PBMC
D.
TIM-4-L gMFI
E.
ns
F.
G.
Bone Marrow
Secondary only
TIM-4 stained
Healthy
AML
TIM-4-L

Figure 1 - Measurement of cell surface TIM-4-L on hematologic malignancies. Cryopreserved bone marrow or PBMC from treatment-naive donors were tested for cell surface TIM-4-L. using flow cytometry. (A) TIM-4-L expression on healthy or AML bone marrow mononuclear cells was measured on live, CD45 intermediate cells for patient samples, or live, CD45 high cells for healthy donors. Samples were measured in duplicate. Average +/- SD is shown, n = 8 for healthy samples, 31 for AML samples. (B) TIM-4-L expression on healthy or AML PBMCs was measured as above. Average +/- SD is shown, n = 4 for healthy samples, 9 for AML samples. (C) TIM-4-L expression on healthy, MCL, or CLL B cells was measured in live, CD45+ CD3- CD19+ cells. Average +/- SD is shown, n = 12 for healthy samples, 3 for MCL samples, 13 for CLL samples. (D) gMFI of TIM-4-L stain was measured in AML BMMCs as above. (E) gMFI of TIM-4-L stain was measured in AML PBMCs as above. (F) gMFI of TIM-4-L was measured in MCL and CLL live, CD45+ CD3- CD19+ cells as above. (G) Example flow plots for TIM-4-L staining of AML or healthy donor BMMC. Data was analyzed using unpaired t-tests for AML samples or one-way ANOVA for MCL and CLL samples, * = p<0.05, ** = p<0.01, **** = p<0.0001. Black dots indicate samples from pre-treated patients.

FIGURE 2 CER-1236 T cells have high chimeric receptor expression, favorable memory profile, and TIM-4-L dose-dependent responses

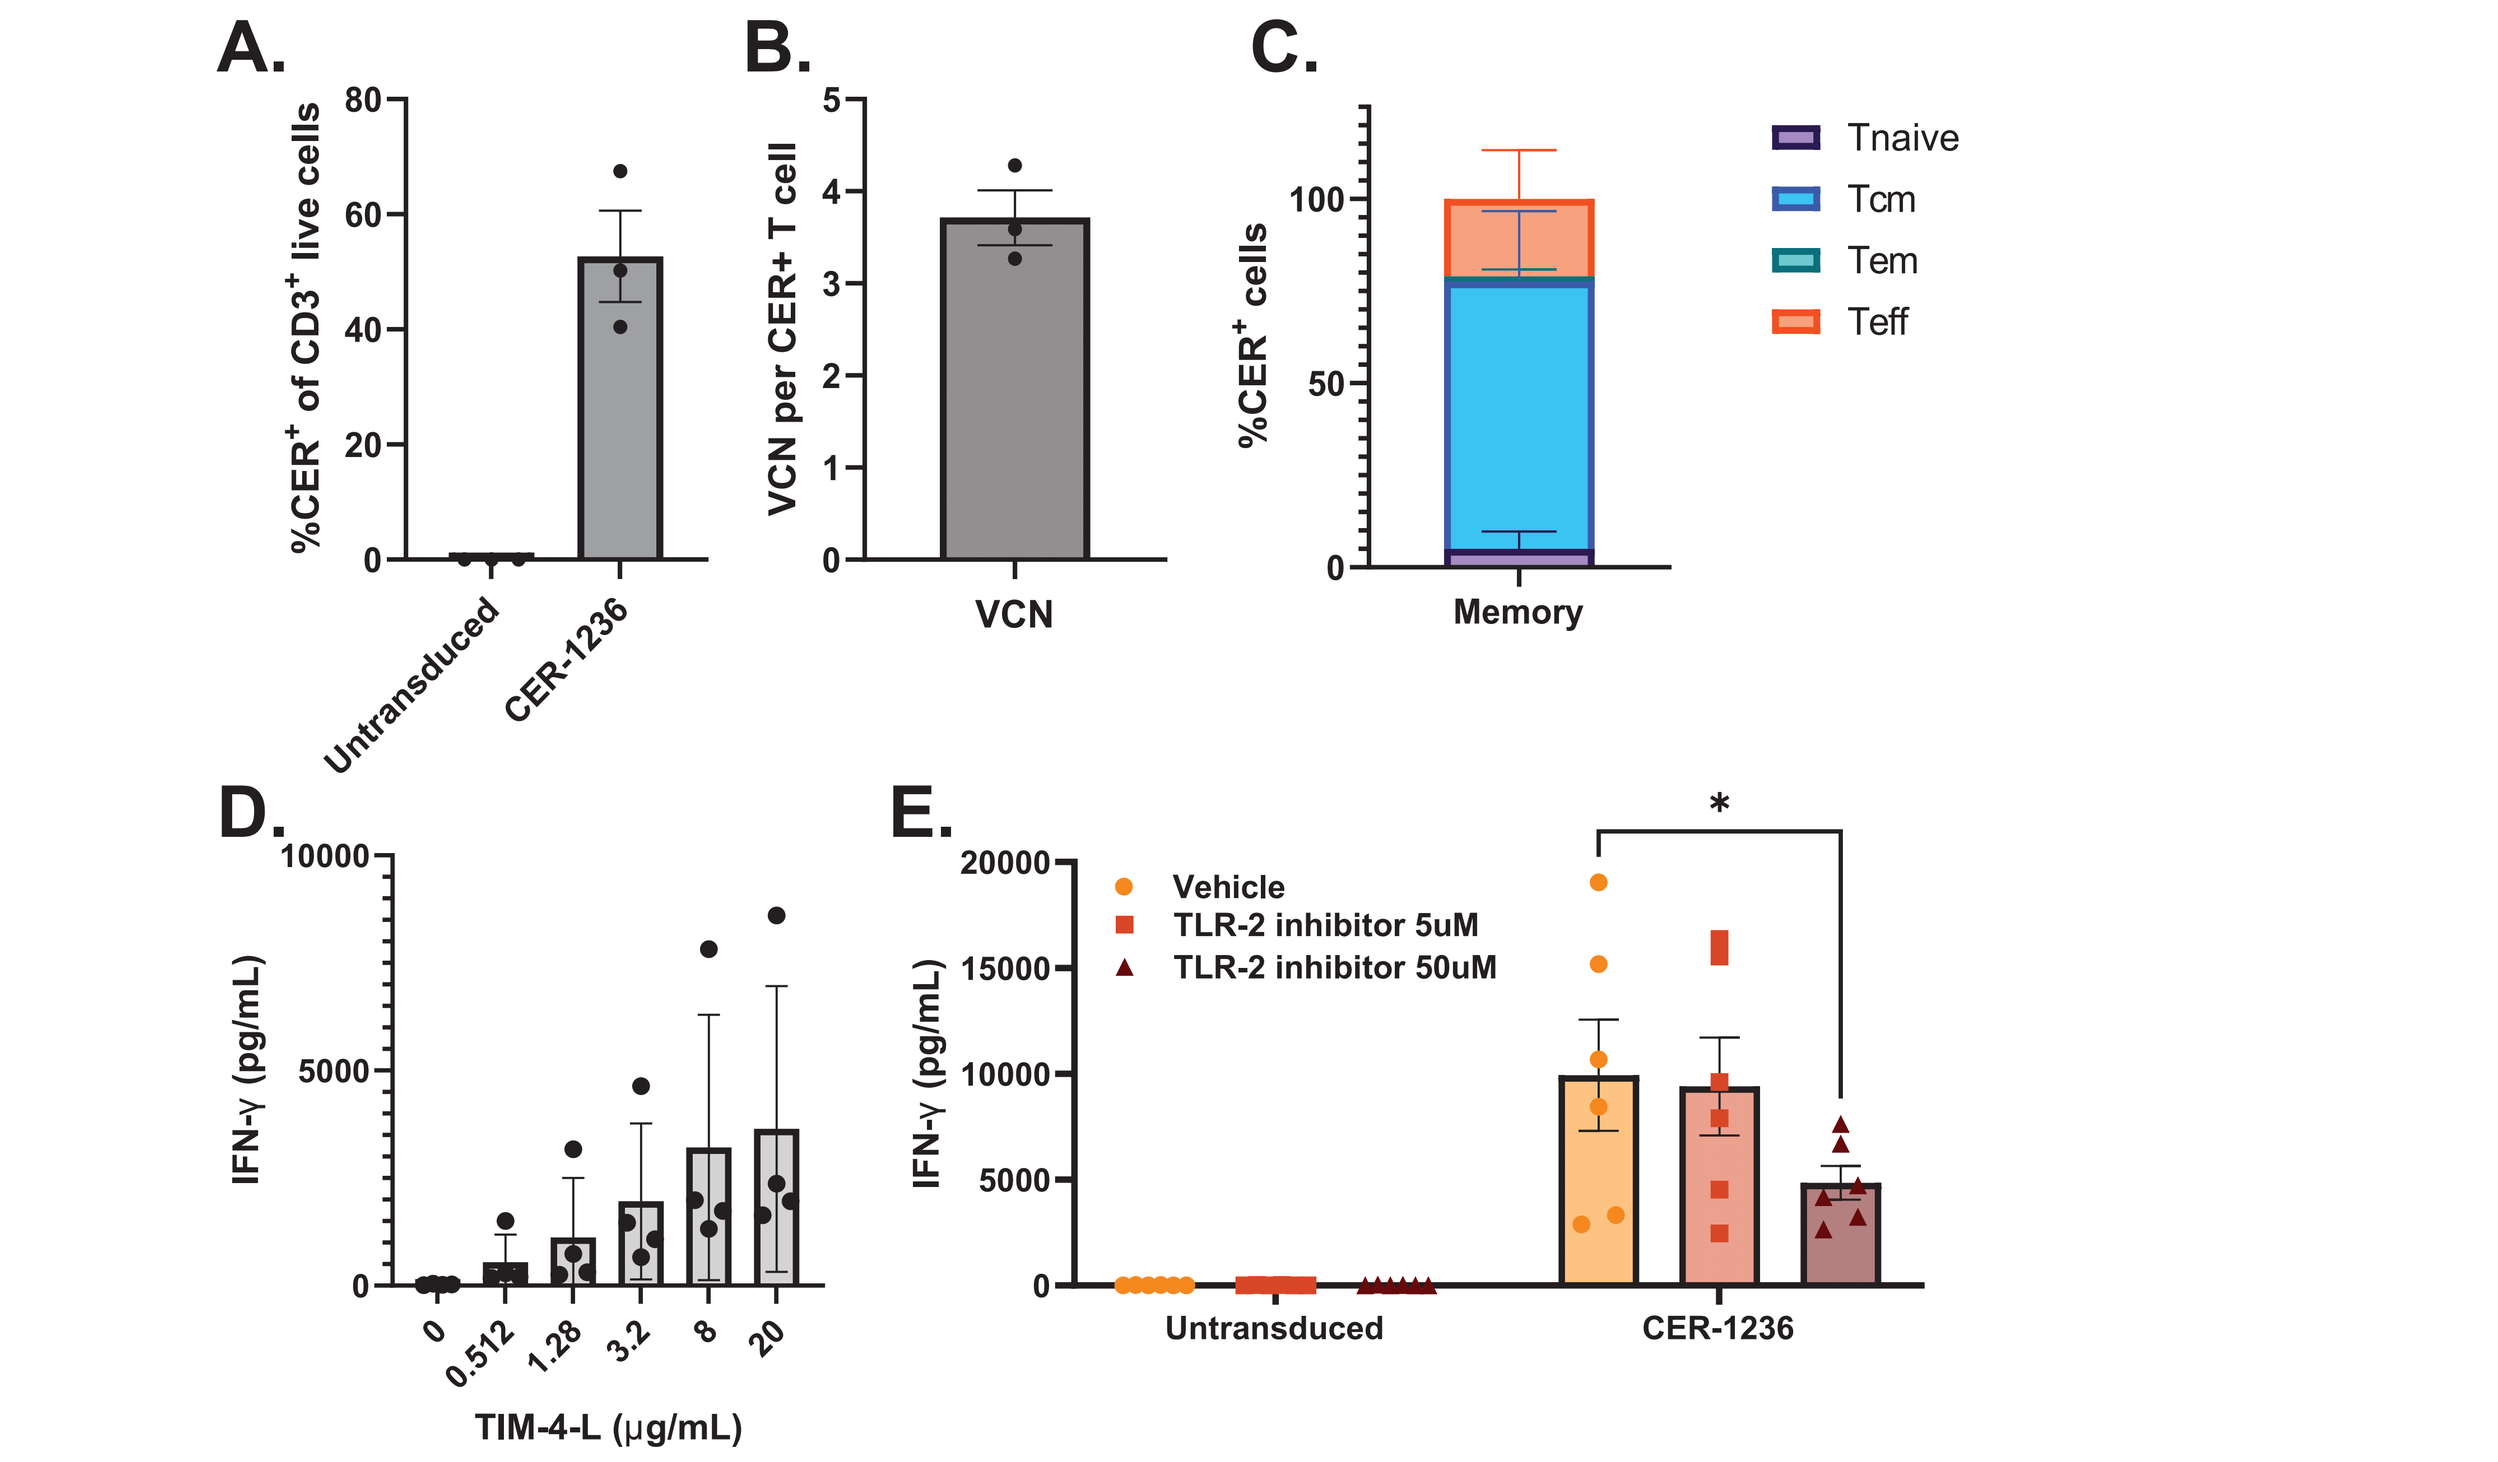
A.
%CER+ of CD3+ live cells
Untransduced
CER-1236
B.
VCN per CER+ T cell
VCN
C.
%CER+ cells
Memory
Tnaive
Tcm
Tem
Teff
D.
IFN-γ (pg/mL)
TIM-4-L (µg/mL)
E.
Vehicle
TLR-2 inhibitor 5uM
TLR-2 inhibitor 50uM
Untransduced
CER-1236

Figure 2 - Characterization of CER-1236 product attributres and target-specific cytokine secretion. CER-1236 T cells were generated from healthy donor leukopaks using a CliniMACS Prodigyand characterized after a 7 day production. (A) CER-1236 transduction efficiency was measured by cell surface expression of the CER-1236 protein. Average +/- SD is shown, n = 3. (B) Vector copy number (VCN) was determined by ddPCR. Average +/- SD is shown, n = 3. (C) CER-1236 T cell memory phenotype was assessed by flow cytometry in live, CD3+ TIM-4+ cells using CCR7 and CD45RA. Tnaive = CCR7+CD45RA+, Tcm = CCR7+CD45RA-, Tem = CCR7- CD45RA-, Teff = CCR7-CD45RA+. Average +/- SD is shown, n = 3. (D) CER-1236 T cells were stimulated using plate-bound TIM-4-L. IFN-γ secretion was measured after 24 hours. Average +/- SD is shown, n = 4. (E) The contribution of TLR2 signaling to CER-1236 T cell response to TIM-4-L was tested by treatment with C29, a small molecule TLR-2 inhibitor. IFN-γ secretion was measured after 24 hours. Average +/- SD is shown, n = 3 with duplicates shown.

FIGURE 3 CER-1236 T cells engulf TIM-4-L+ targets and can cross present antigen to bystander T cells

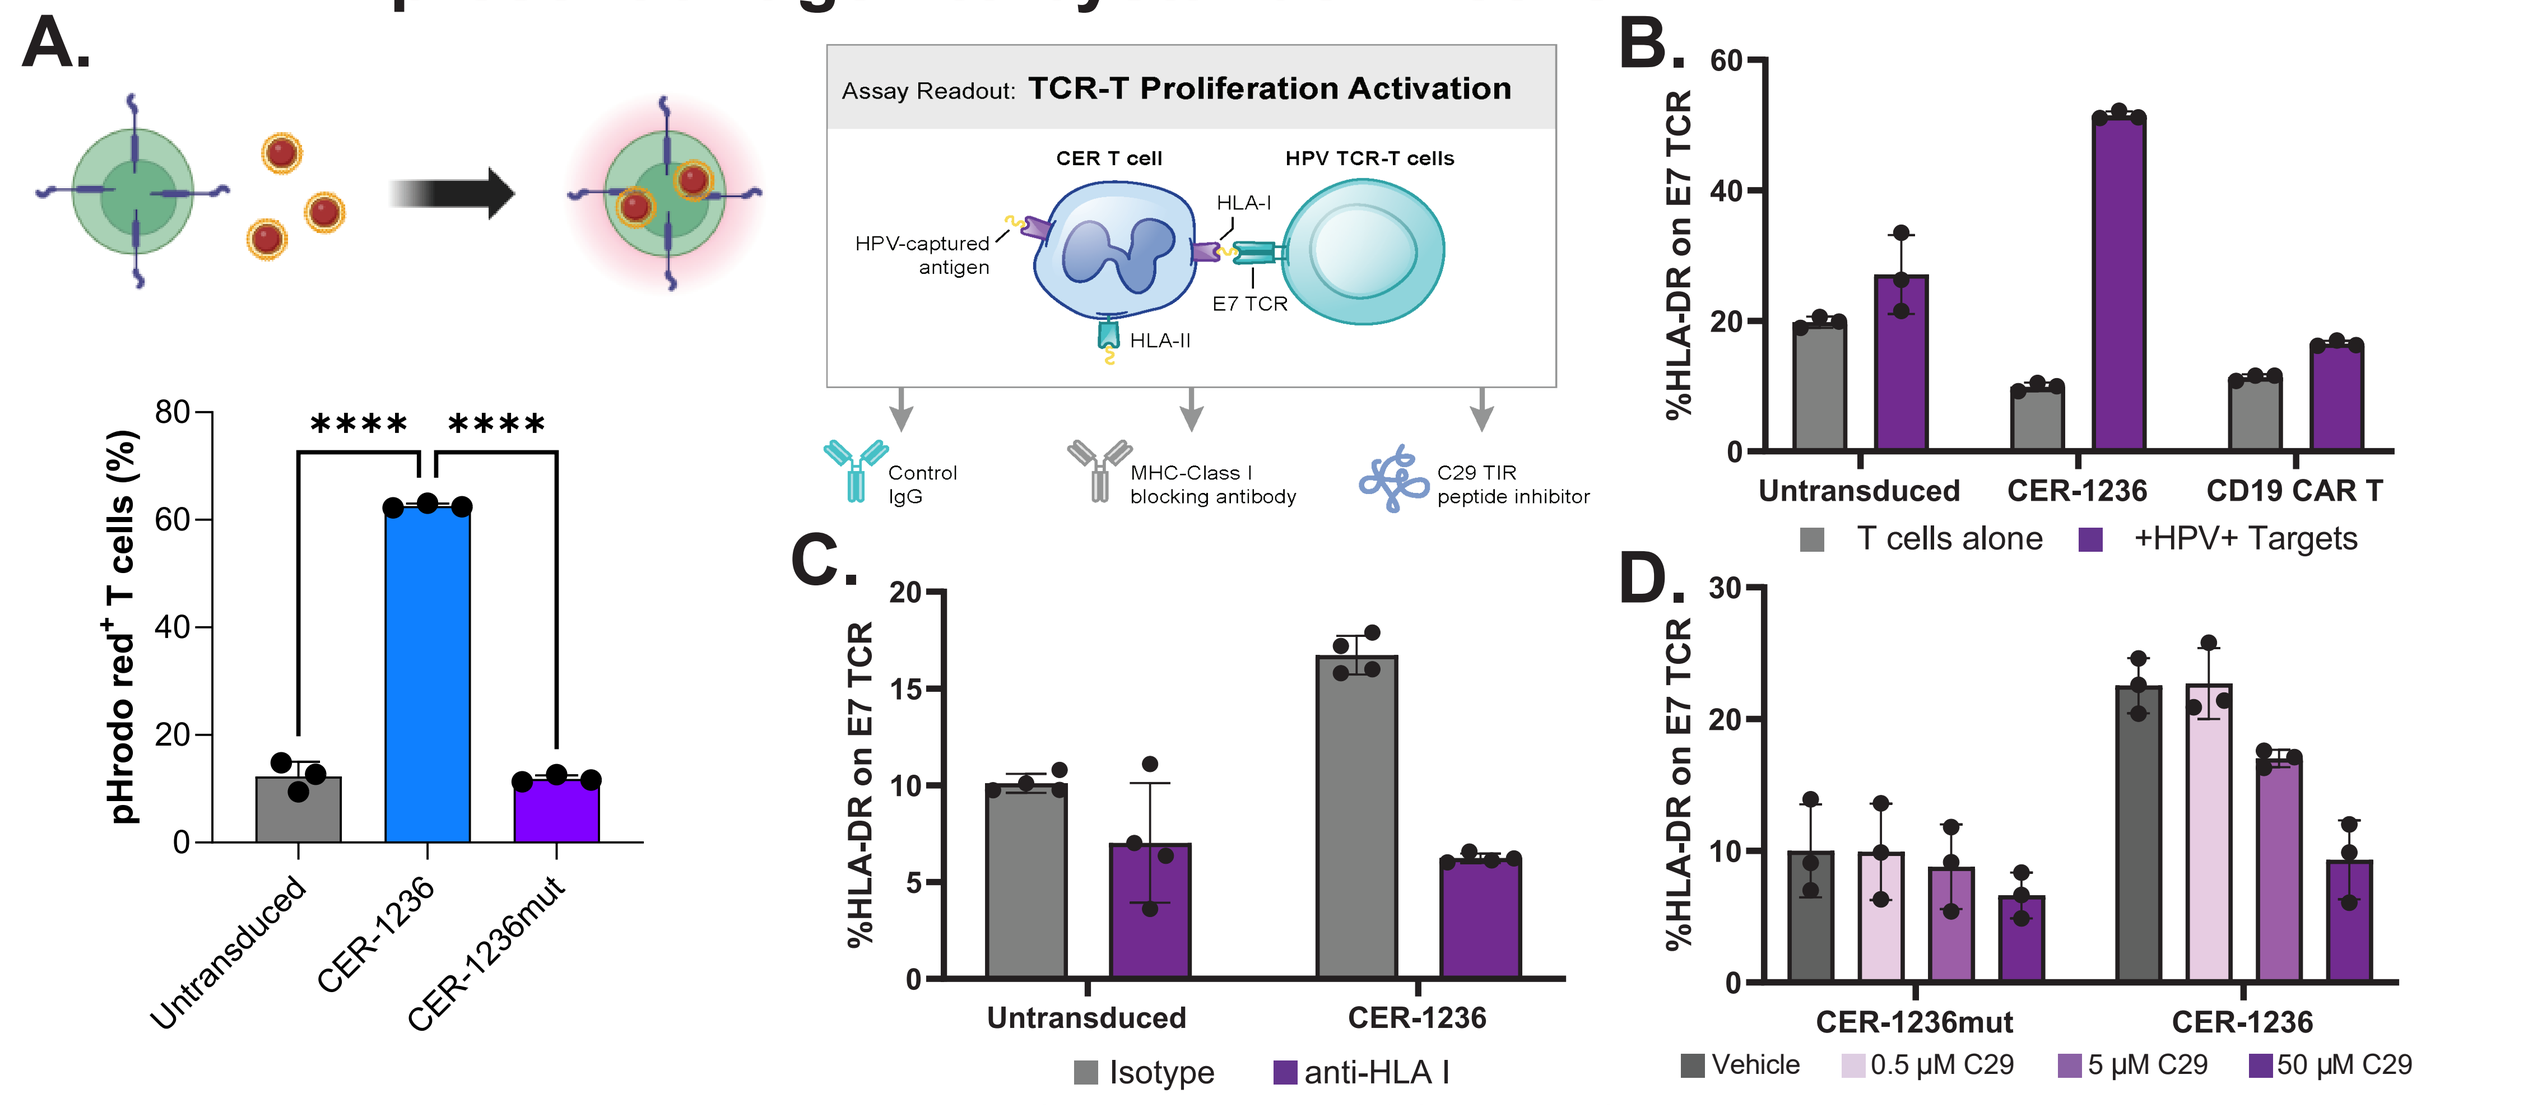
A.
Assay Readout: TCR-T Proliferation Activation
CER T cell
HPV TCR-T cells
HPV-captured antigen
HLA-I
E7 TCR
HLA-II
Control IgG
MHC-Class I blocking antibody
C29 TIR peptide inhibitor
pHrodo red+ T cells (%)
Untransduced
CER-1236
CER-1236mut
B.
%HLA-DR on E7 TCR
CD19 CAR T
T cells alone
+HPV+ Targets
C.
Isotype
anti-HLA I
D.
CER-1236mut
Vehicle
0.5 µM C29
5 µM C29
50 µM C29

Figure 3 - Phagocytosis and antigen cross-presentation by CER-1236 T cells. (A) CER-1236, CER-1236 binding site mutant, or untransduced T cells were co-cultured with TIM-4-L-coated pHrodo-Red 0.5 µm beads for 24h. Cells were washed to remove exterior beads, and bead engulfment was measured by flow cytometry. Average +/- SD is shown, n = 3 triplicate samples. Significance was tested by one-way ANOVA with Tukey's post test. **** = p<0.0001. (B) CER-1236, untransduced, or a CD19 CAR T cells were co-cultured with TIM-4-L+ E7+ SCC152 cells for 48h. T cells were isolated from co-culture and then co-cultured with autologous E7 TCR+ T cells for 72h. Expression of the activation marker HLA-DR on E7 TCR+ T cells was measured by flow cytometry. Average +/- SD is shown, n = 3 triplicate samples. (C) CER-1236 or untransduced T cells were cultured as above in the presence of HLA class I blocking antibody or isotype control. Average +/- SD is shown, n = 4 replicate samples. (D) CER-1236 or CER-1236 binding site mutant T cells were cultured as above in the presence of the TLR2 inhibitor C29 . Average +/- SD is shown, n = 4 replicate samples.

FIGURE 4 CER-1236 T cells eliminate AML cell lines positive for TIM-4-L

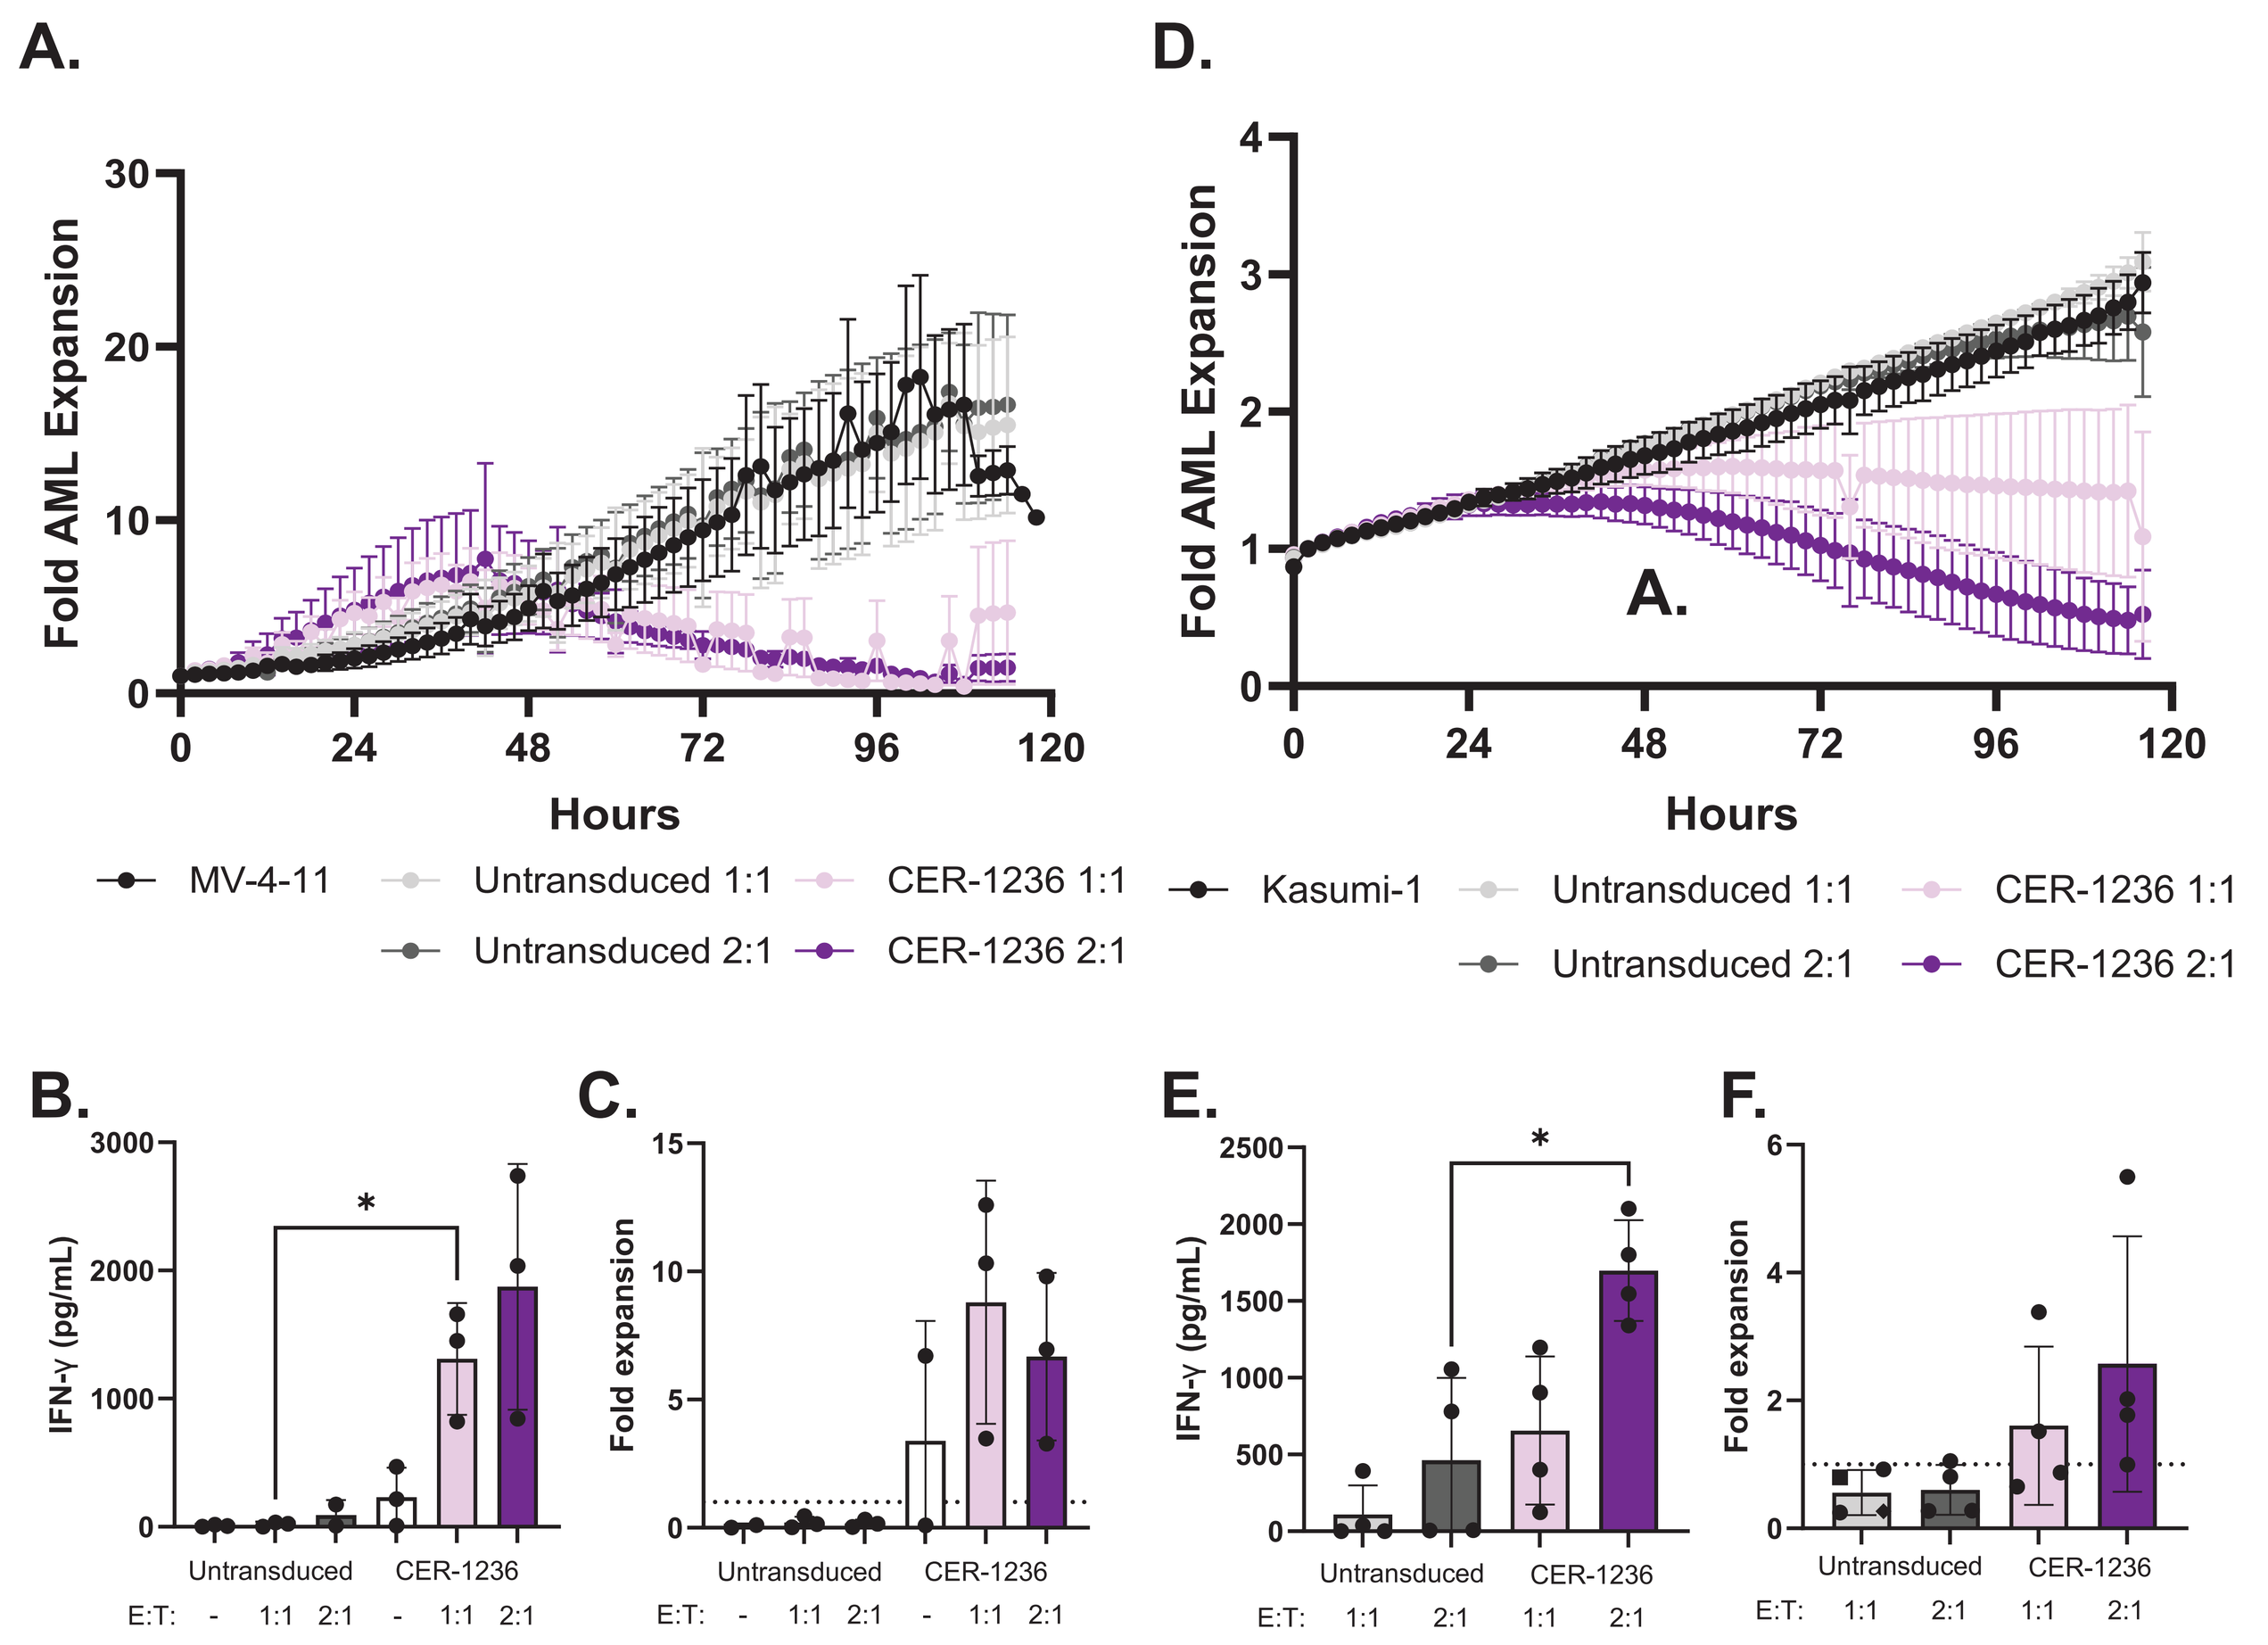
A.
Fold AML Expansion
Hours
MV-4-11
Untransduced 1:1
Untransduced 2:1
CER-1236 1:1
CER-1236 2:1
D.
Kasumi-1
B.
IFN-γ (pg/mL)
C.
Fold expansion
E.
F.
Untransduced
CER-1236
E:T:

Figure 4 - Cytotoxic and effector functions of CER-1236 T cells against AML. (A) CER-1236 or untransduced T cells were co-cultured with the MV-4-11 AML cell line engineered to express mCherry at the indicated E:T ratios and cytotoxicity was assessed by incucyte. Expansion of MV-4-11 cells is shown. Average +/- SEM is shown, N = 3 donors. (B) Secretion of IFN-γ after 5 days of co-culture was measured by ELISA. Average +/- SD is shown, n = 3. Significance was determined by paired t-test. * = p<0.05. (C) T cell expansion at the indicated E:T ratios was measured after 5 days of co-culture by flow cytometry. Average +/- SD is shown, n = 3 for co-culture, n = 2 for T cell alone. (D) CER-1236 or untransduced T cells were co-cultured with the Kasumi-1 AML cell line engineered to express mCherry at the indicated E:T ratios and cytotoxicity was assessed by incucyte. Expansion of Kasumi-1 cells is shown. Average +/- SEM is shown, N = 3 donors. (E) Secretion of IFN-γ after 5 days of co-culture was measured by ELISA. Average +/- SD is shown, n = 4. Significance was determined by paired t-test. * = p<0.05. (F) T cell expansion was measured after 5 days of co-culture by flow cytometry. Average +/- SD is shown, n = 4.

FIGURE 5 CER-1236 T cells eliminate AML xenografts in vivo

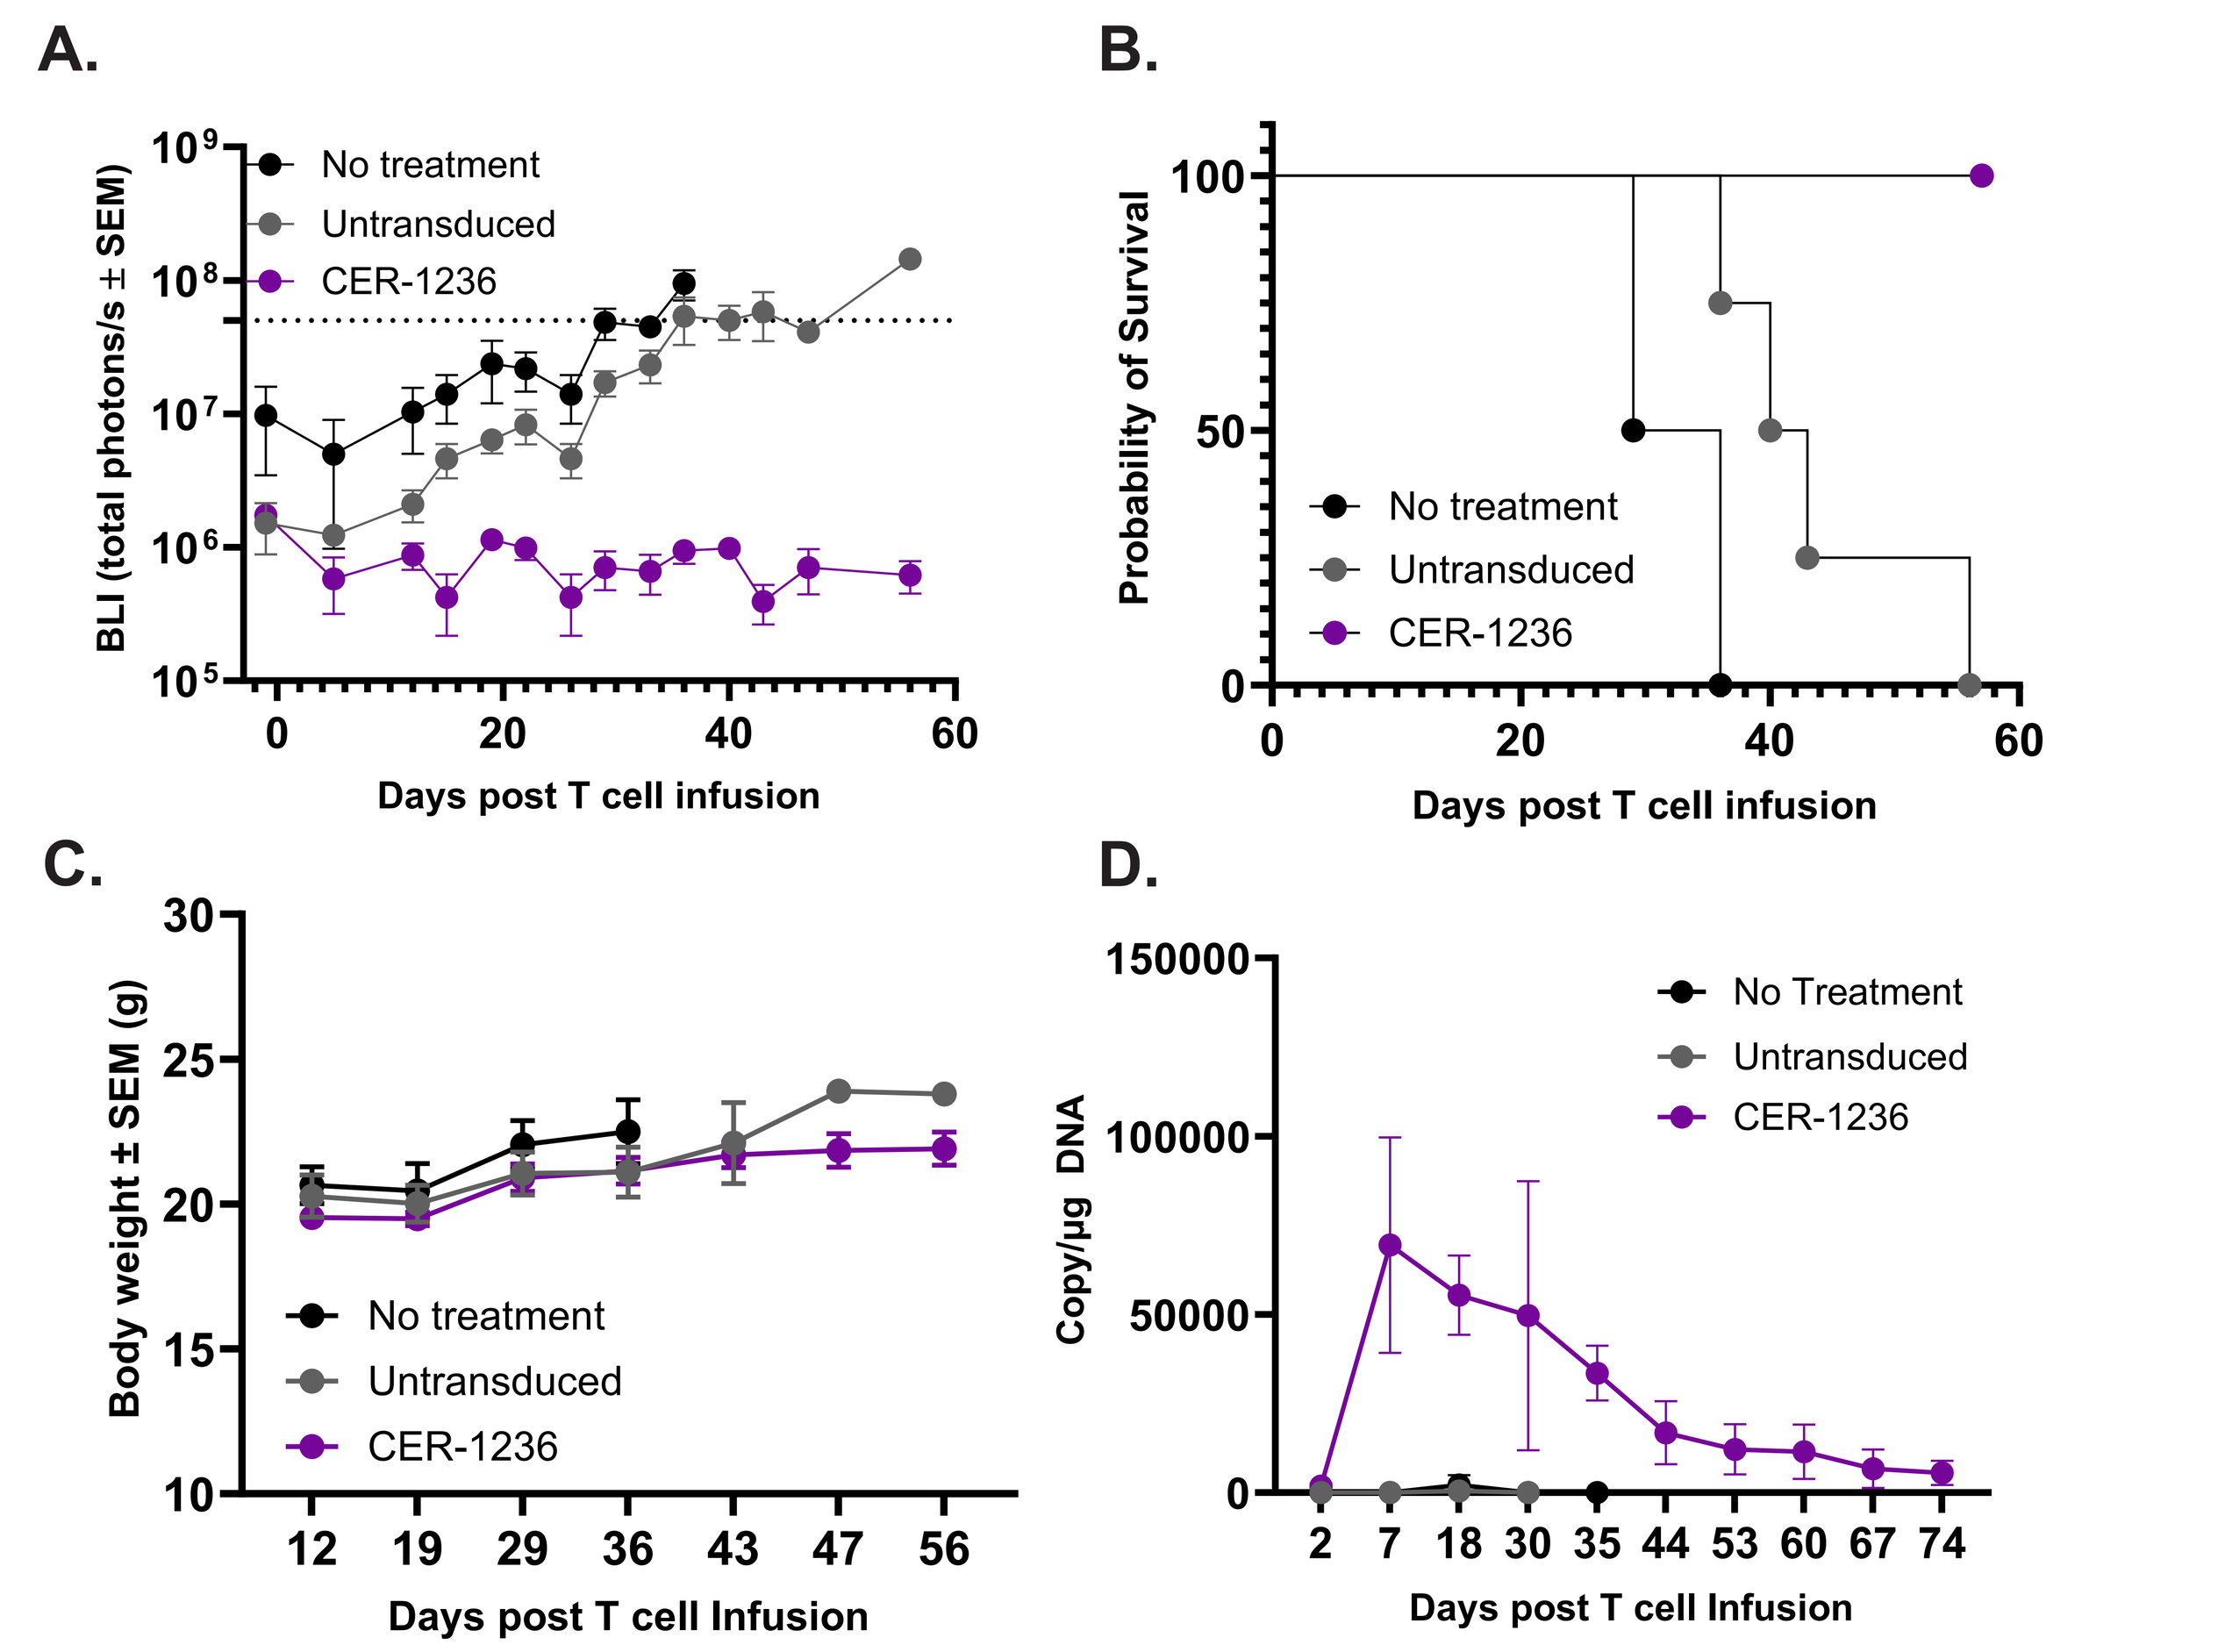
A.
BLI (total photons/s ± SEM)
No treatment
Untransduced
CER-1236
Days post T cell infusion
B.
Probability of Survival
C.
Body weight ± SEM (g)
Days post T cell Infusion
D.
Copy/µg DNA
No Treatment

Figure 5 - CER-1236 mediates anti-tumor effect against Kasumi-1 AML cell in vivo. NSG MHC dKO mice were engrafted i.v. with 2e6 GFP/fLuc+ Kasumi-1 cells for 10d. Mice were treated with 7.5e6 CER-1236+ T cells or an equivalent number of untransduced T cells. (A) Tumor burden was monitored by BLI imaging. Mice were removed from study when BLI exceeded 5e7 photons/s. Average +/- SEM is shown, n=6 for CER-1236, n = 4 for untransduced and no treatment. (B) Survival of Kasumi-1 engrafted mice treated with CER-1236 T cells or control cells. (C) Body weight was measured to monitor overall health. Average +/- SEM is shown. (D) CER-1236 T cell expansion in the blood was measured by ddPCR. Copies per µg DNA was calculated using the equation: (Copies CER-1236*100000)/(Copies HAT*3.3).

FIGURE 6 TIM-4-L expression is specific to malignant cells and not normal bone marrow or HSPCs

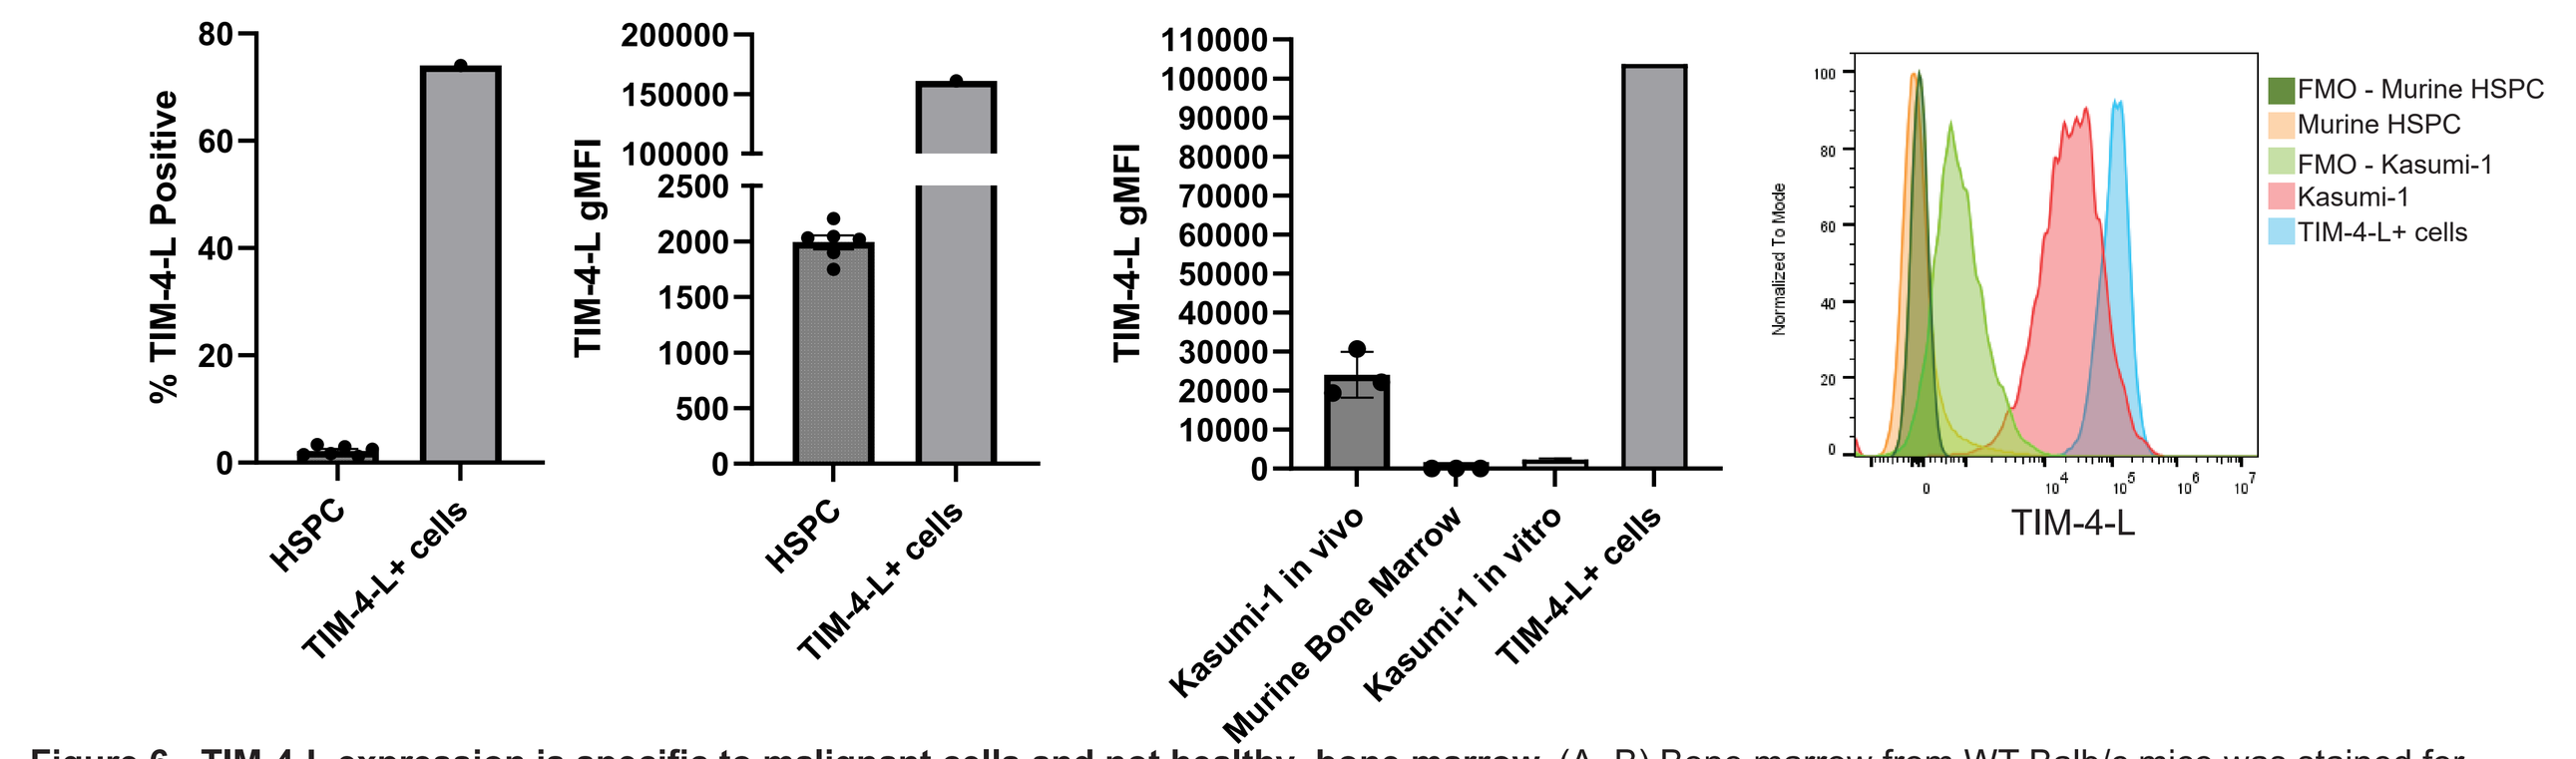
% TIM-4-L Positive
HSPC
TIM-4-L+ cells
TIM-4-L gMFI
Kasumi-1 in vivo
Murine Bone Marrow
Kasumi-1 in vitro
FMO - Murine HSPC
Murine HSPC
FMO - Kasumi-1
Kasumi-1
TIM-4-L+ cells
TIM-4-L

Figure 6 - TIM-4-L expression is specific to malignant cells and not healthy bone marrow. (A, B) Bone marrow from WT Balb/c mice was stained for TIM-4-L on hematopoietic stem and progenitor cells (HSPC) . Percent TIM-4-L positive cells (A) and per-cell TIM-4-L (B) was assessed in live CD45+EP-CR+CD48-CD150+ cells by flow cytometry. TIM-4-L positive cells are shown for reference. Average +/- SD is shown, n = 6 animals. (C) NSG MHC dKO mice were engrafted with 2e6 Kasumi-1 GFP/fLuc cells i.v. for 70d. Bone marrow was stained for TIM-4-L and assessed by flow cytometry. TIM-4-L was measured on hCD45+GFP+ Kasumi-1 cells, or healthy hCD45-GFP- murine bone marrow, TIM-4-L positive cells and in vitro Kasumi-1 cells are shown for reference. Average +/- SD is shown, n = 3 mice for bone marrow samples, n = 1 for reference cells. (D) Example histograms of TIM-4-L staining.

FIGURE 7 CER-1236 T cell secreted effectors can enhance TIM-4-L expression on AML cells

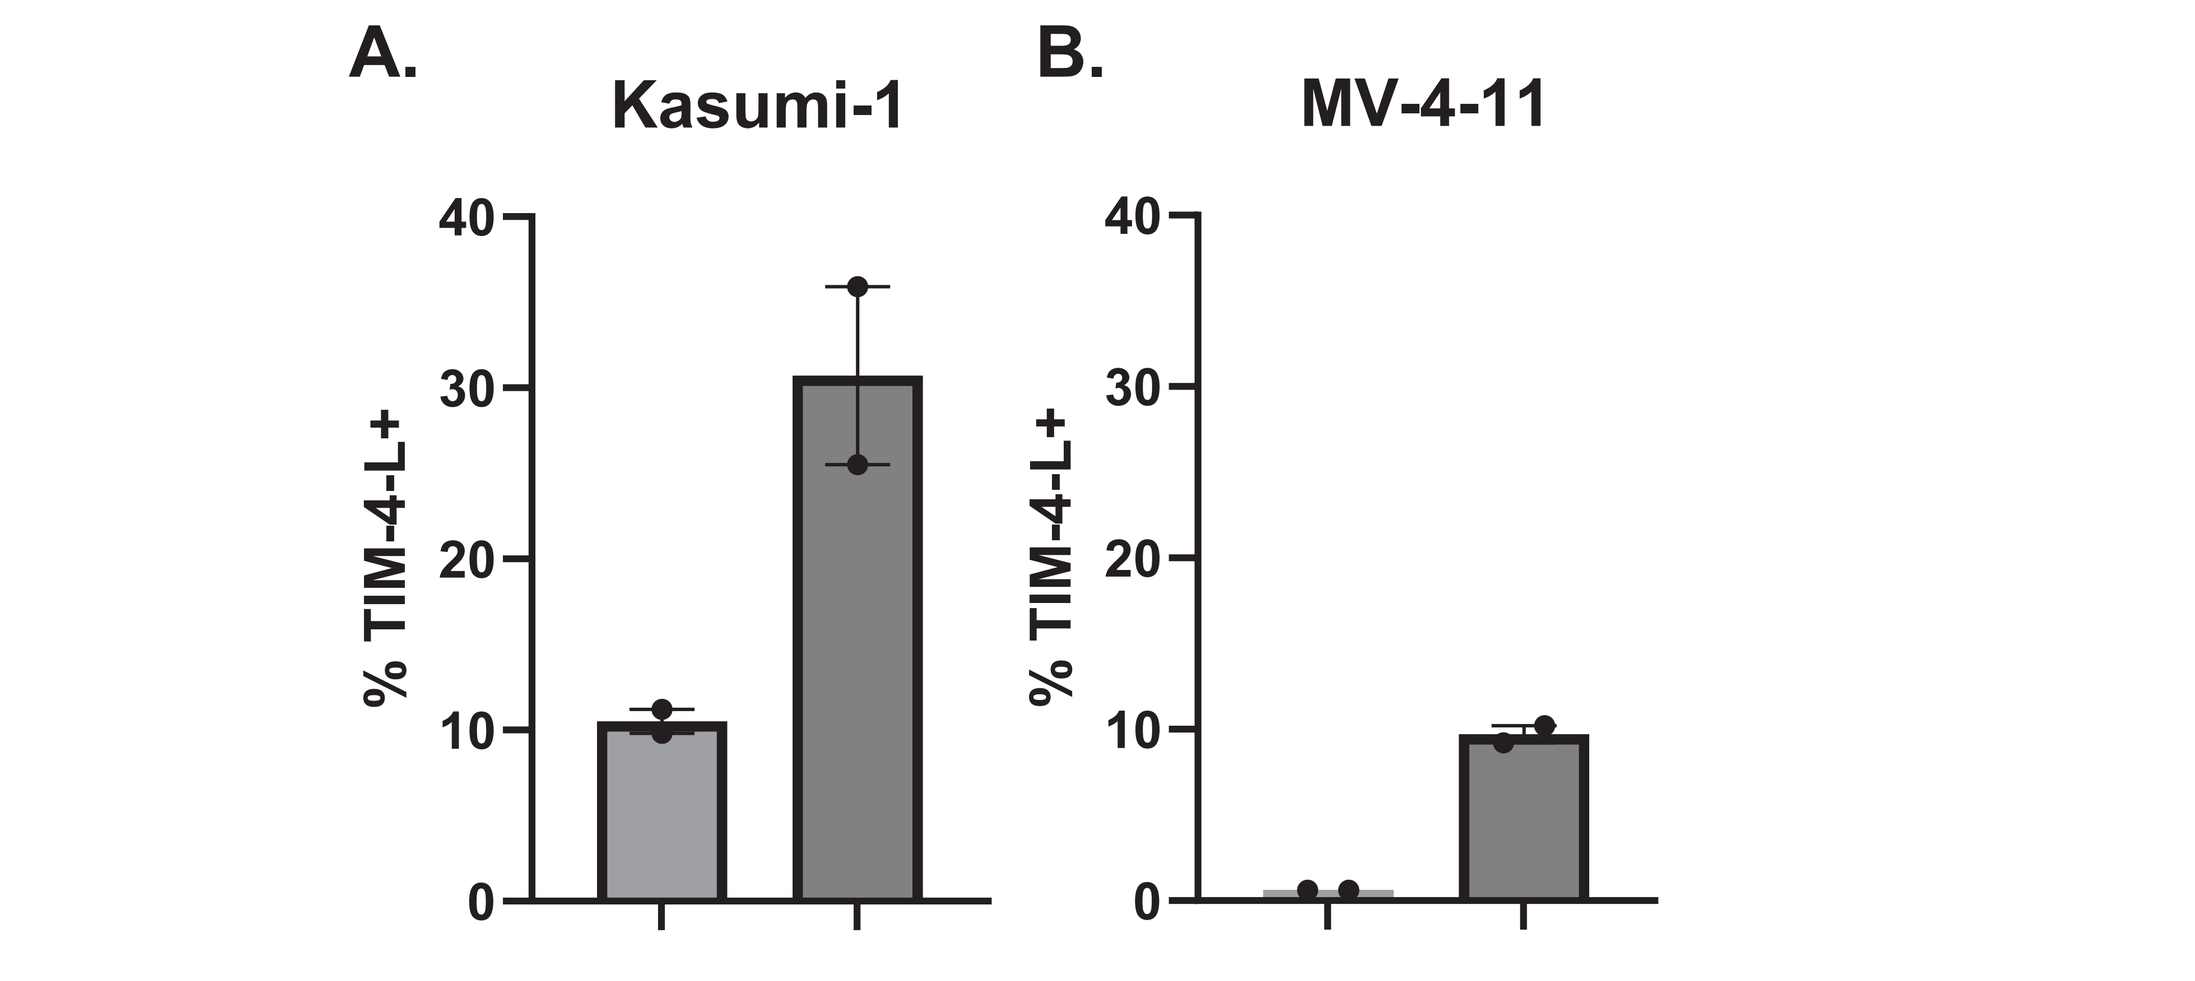
A.
Kasumi-1
B.
MV-4-11
% TIM-4-L+
Cultured Supernatant:

Figure 7 - CER-1236 T cell secreted factors induce TIM-4-L expression on AML. CER-1236 T cells were stimulated with 5 µg/ml plate-bound TIM-4-L for 24h. Supernatant was collected and spun down to remove any remaining T cells, then frozen. (A) Kasumi-1 or (B) MV-4-11 AML cells were cultured in CER-1236 T cell conditioned media or fresh media for 48h. After culture, AML cells were resuspended and stained for TIM-4-L expression by flow cytometry using recombinant human TIM-4. TIM-4-L positivity is shown in live cells. Average +/- range is shown, n = 2.

CER-1236 Mechanism of Action

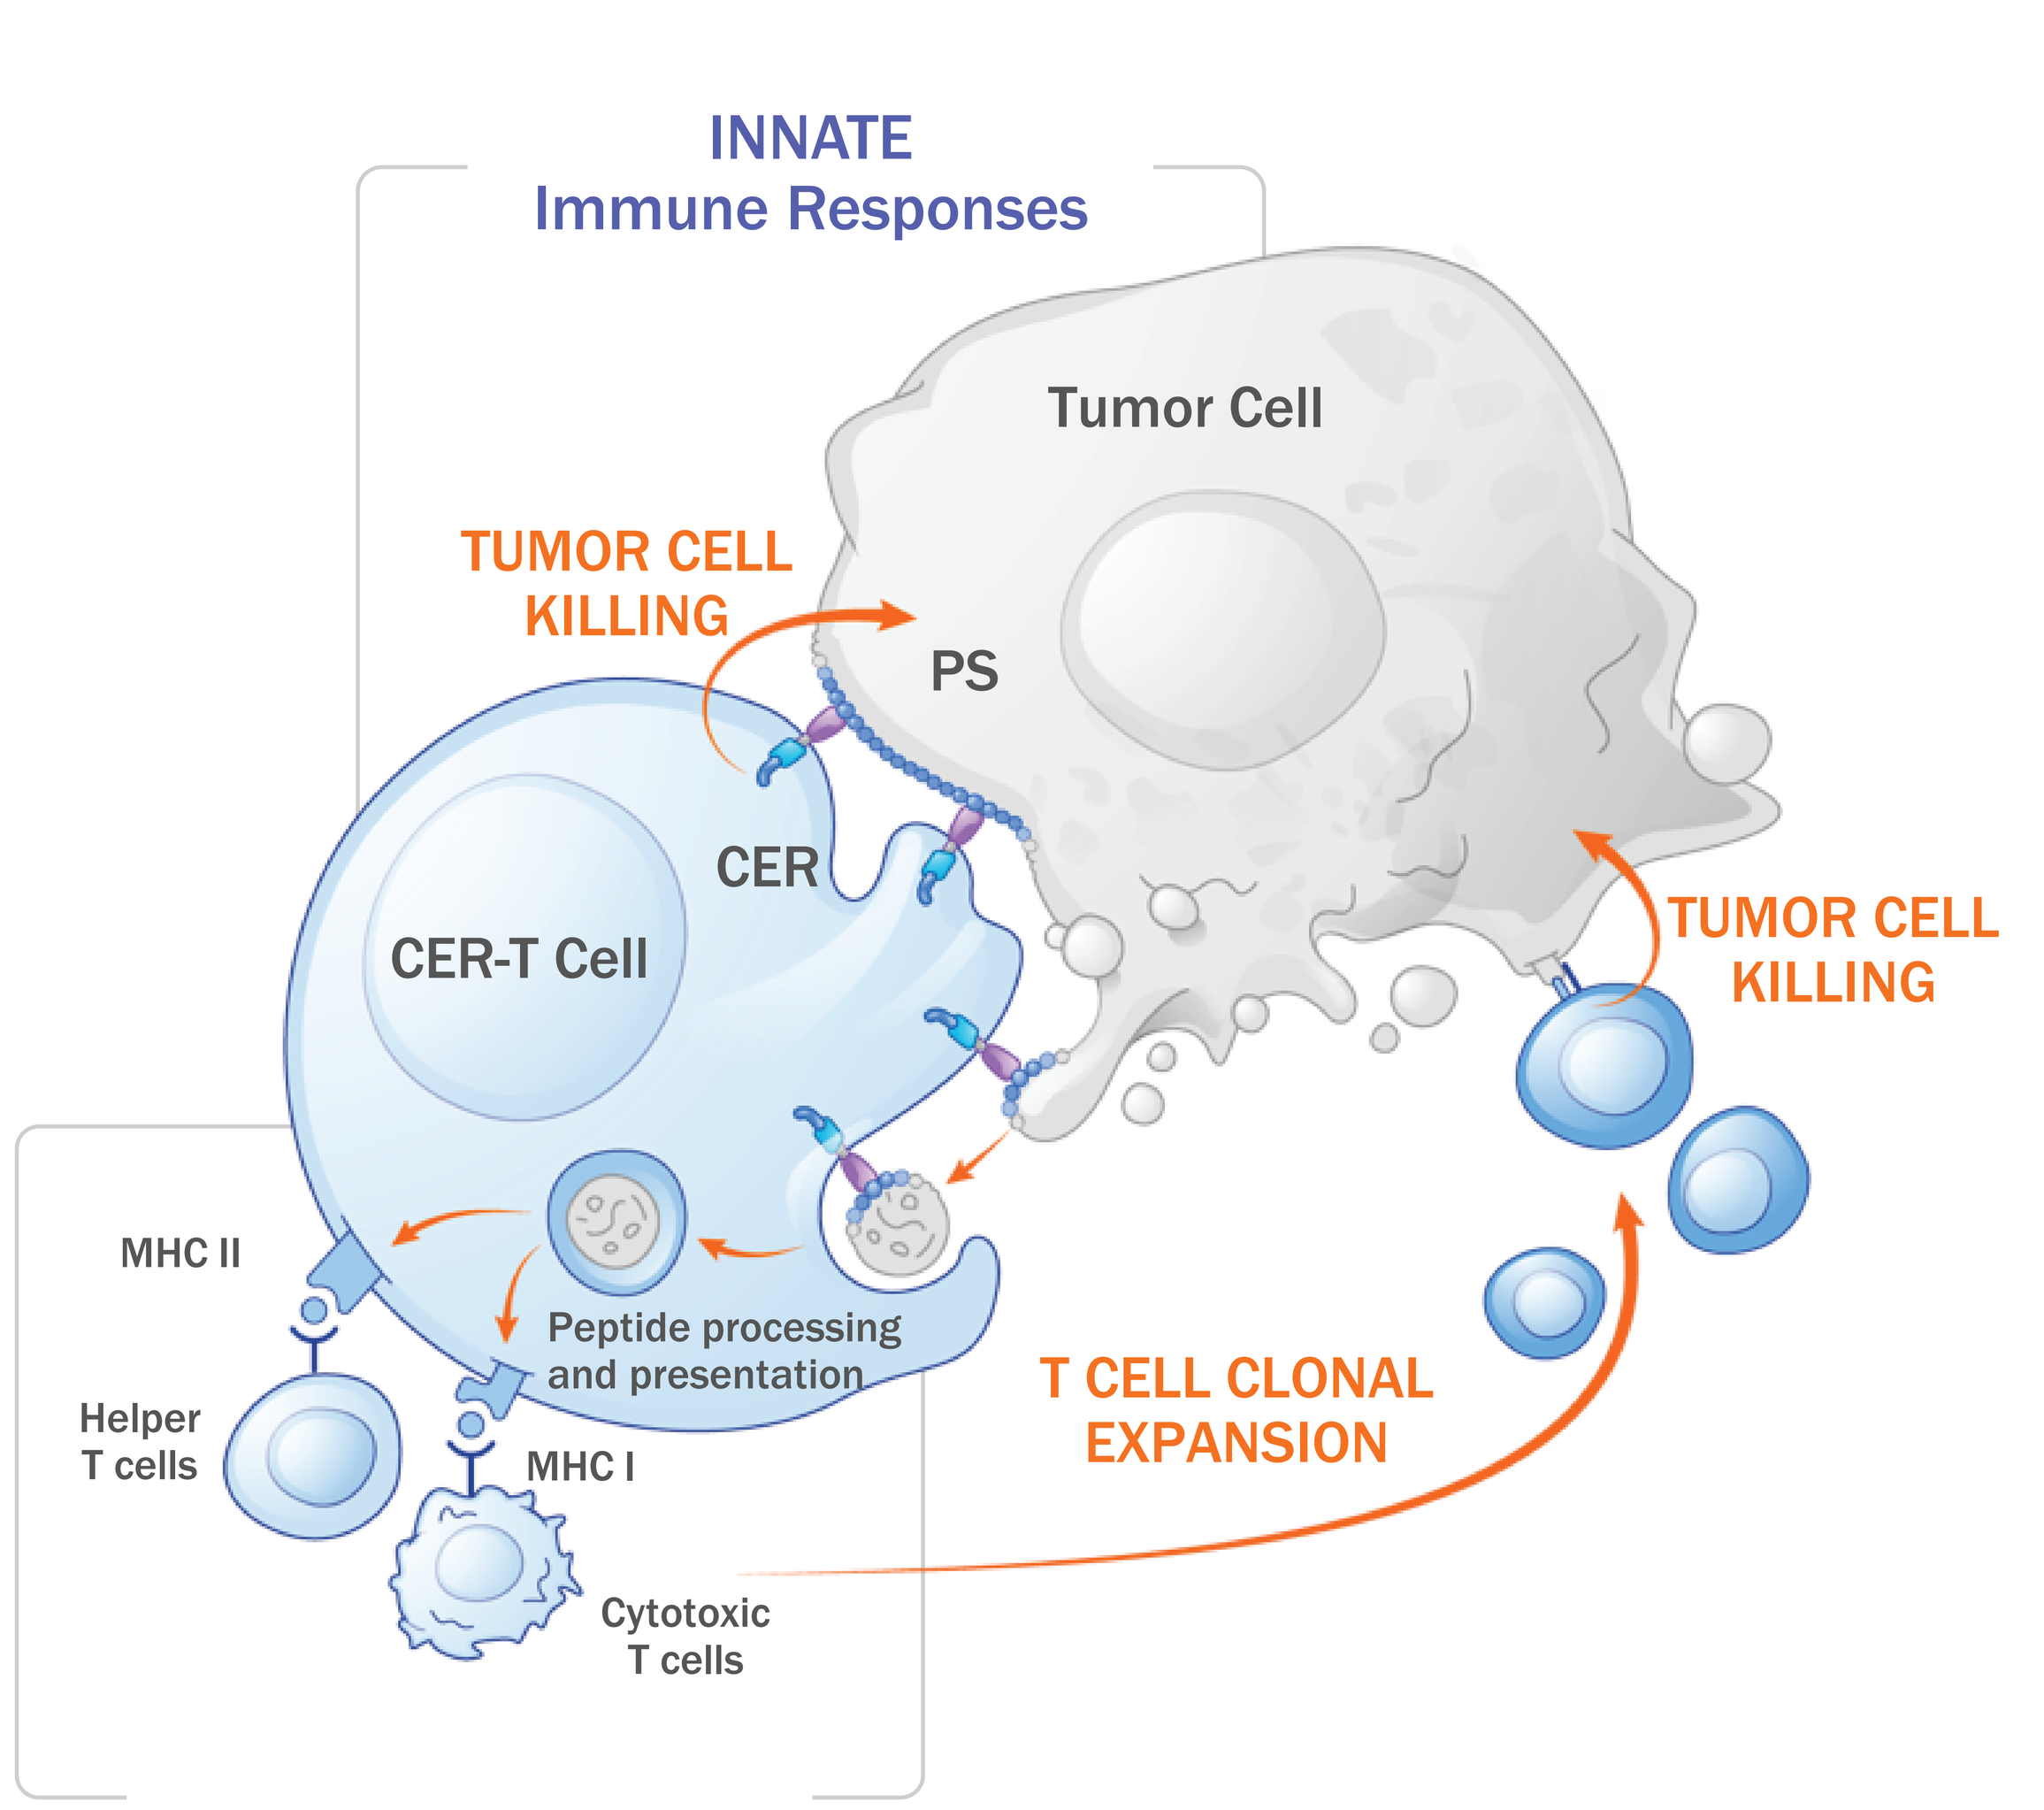
INNATE
Immune Responses
Tumor Cell
TUMOR CELL KILLING
PS
CER
CER-T Cell
TUMOR CELL KILLING
MHC II
Helper T cells
Peptide processing and presentation
MHC I
Cytotoxic T cells
T CELL CLONAL EXPANSION

Conclusions

- Aberrant TIM-4-L expression is observed on malignant cells of multiple lineages, but minimally on healthy cells including bone marrow and HSPCs.
- CER-1236 T cells engineered to recognize TIM-4-L can be highly transduced, have a favorable memory phenotype, and a dose-dependent response to TIM-4-L.
- CER-1236's unique extracellular domain and intracellular signaling domains impart the capacity to phagocytose and cross-present antigen from targeted cells.
- CER-1236 T cells show cytotoxic function, secrete effector cytokines, and proliferate in response to AML cell lines in vitro.
- TIM-4-L expression can be further enhanced by CER-1236 secreted factors, potentially enhancing effector functions.

Acknowledgements

We would like to acknowledge CERo Therapeutics employees past and present who contributed to this work.

Disclosures

BC, AB, EO, ZY, BY, and HN are all employees of CERo Therapeutics Holdings, Inc. DC is an employee and reports ownership of CERo Therapeutics Holdings, Inc.